Evaluating the Use of Merlin-YAP Dual-label Immunohistochemistry for Predicting Response to TEAD Inhibitor VT3989

Tracy T. Tang[1], Kelly J. Gordon[2], Sebastian Müller[3], Yufeng Li[1], Jacob Lewis[4], Christine Hale[4], Timothy Cloherty[4], Noreen McBrearty[4], Neelesh Sharma[1], and Leonard Post[1]

[1]Vivace Therapeutics, Inc., [2]Boudicca Dx. LLC, [3]Alva Genomics, [4]Discovery Life Sciences

Abstract #207

Background

- TEAD transcription factors are the major effectors of the Hippo-YAP/TAZ pathway involved in cell proliferation, survival, and cell migration[1].
- Genetic alterations of pathway components (e.g., *NF2* which encodes the Merlin protein) leading to constitutive YAP/TAZ nuclear localization and TEAD activation have been reported in a variety of human malignancies[2].
- Preclinical studies demonstrated that TEAD inhibitor VT3989 disrupts YAP/TAZ-TEAD interaction, suppresses TEAD transcriptional activity, and selectively blocks proliferation of *NF2*-deficient mesothelioma *in vitro* and inhibits *NF2*-deficient xenograft tumor growth *in vivo*[3].
- Screening of a large panel of human mesothelioma cell lines showed that cell lines without detected *NF2* mutations also exhibited *in vitro* sensitivity to VT3989. Western blot analysis showed that these sensitive cell lines without detected *NF2* mutations were negative for Merlin protein expression.
- VT3989 is being evaluated in an ongoing phase 1 clinical trial (NCT04665206); partial responses in mesothelioma patients have been demonstrated.
- Responses were observed in patients with loss of function *NF2* mutations, as well as in patients without detected NF2 mutations based on NGS-based local laboratory testing.
- A Merlin-YAP dual immunohistochemistry (IHC) assay has previously been reported[4]. The goal of this study was to develop a similar dual IHC assay to evaluate both Merlin and YAP protein expression and co-localization to provide further biomarker insights on VT3989 clinical responses.

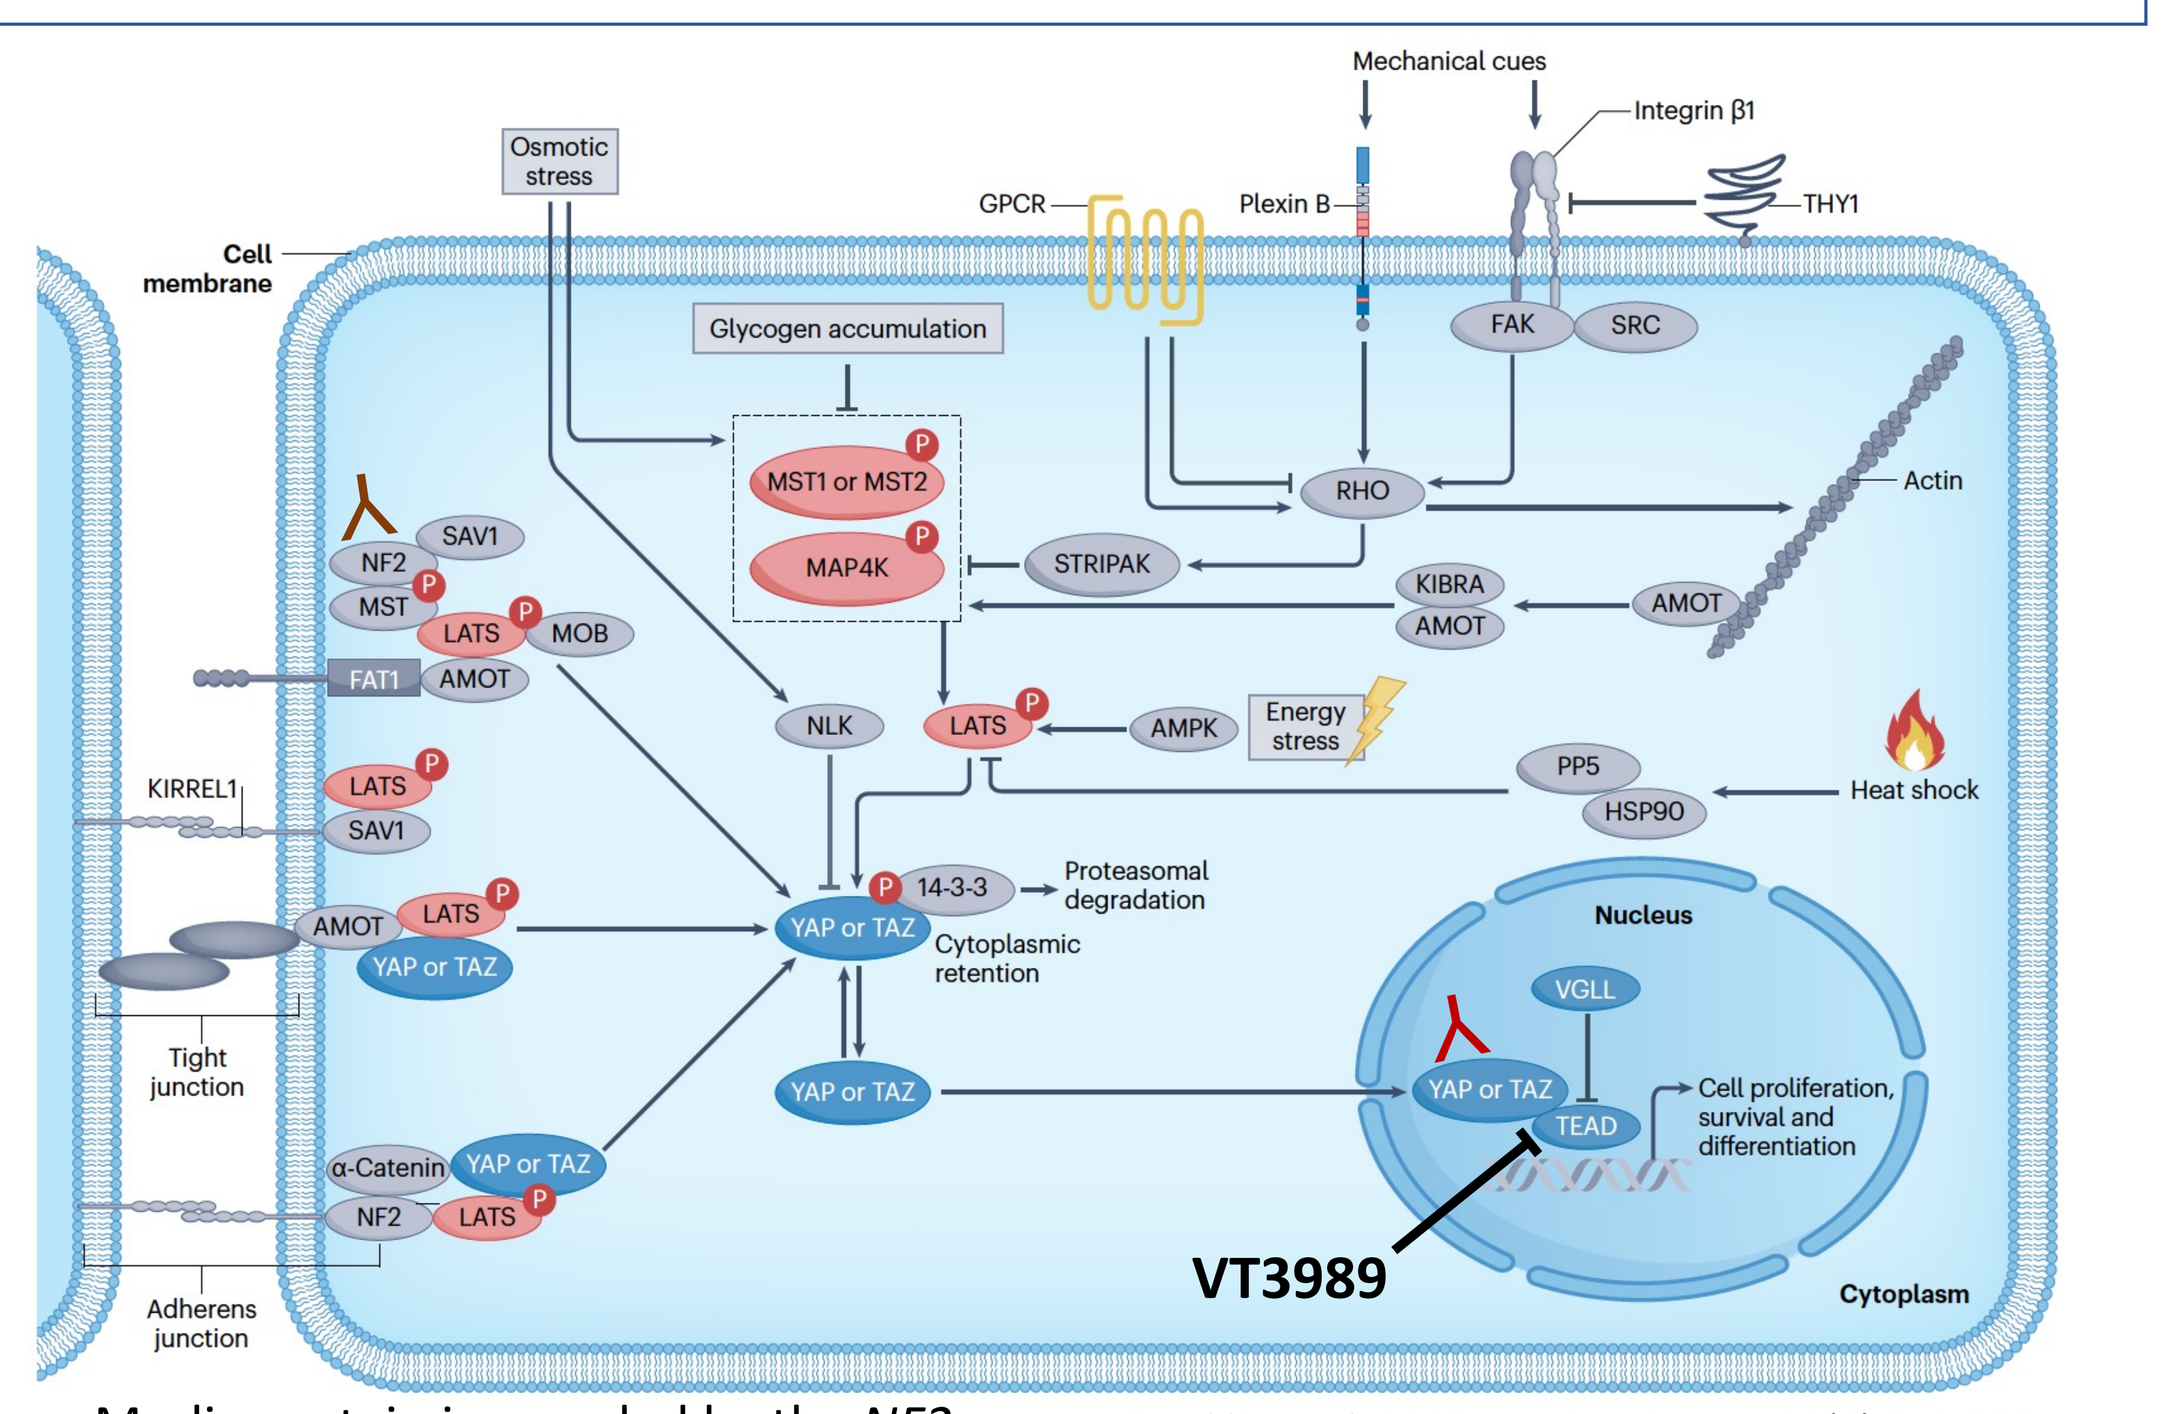

Mechanical cues
Integrin β1
THY1
Osmotic stress
Cell membrane
GPCR
Plexin B
FAK
SRC
Glycogen accumulation
MST1 or MST2
MAP4K
RHO
Actin
STRIPAK
KIBRA
AMOT
SAV1
NF2
MST
LATS
MOB
FAT1
NLK
AMPK
Energy stress
PP5
HSP90
Heat shock
KIRREL1
14-3-3
Proteasomal degradation
YAP or TAZ
Cytoplasmic retention
Nucleus
VGLL
TEAD
Cell proliferation, survival and differentiation
Tight junction
α-Catenin
Adherens junction
VT3989
Cytoplasm

- Merlin protein is encoded by the *NF2* gene. Franklin et al, 2023, Nat Rev Cancer. 23(8):512-525

Summary and Conclusions

- The duplex IHC assay resulted in high specificity for Merlin staining and a visible correlation between Merlin and YAP protein expression. Within the mesothelioma tissues screened, both Merlin and YAP stained as expected, with varying degrees of Merlin cytoplasmic reactivity and YAP cytoplasmic and nuclear reactivity. Although Merlin expression was observed in lymphocytes, only Merlin staining in the tumor cells were evaluated for H-score calculation.
- The Merlin-YAP duplex IHC assay showed that VT3989 activity as measured by partial responses is selective for mesotheliomas with loss of Merlin. This includes identification of Merlin-negative VT3989 partial response mesotheliomas without detected NF2 mutations. Thus, data from this study highlight the importance of comprehensively evaluating pathway components at the genomics (NF2 mutations), transcriptome, and protein level (Merlin & YAP expression) to more fully understand the biomarker landscape of each responder. Further development is ongoing to determine if this Merlin-YAP duplex IHC assay could be applied for patient selection.

VT3989 preclinical activity in Merlin negative mesothelioma without detected *NF2* mutation

A

Cell Line	VT3989 (Multi-TEAD) GI50 (nM)	VT3989 (Multi-TEAD) Max inh %	NF2 Mutation	Merlin Level
NCI-H226	9	101	very low NF2 transcript	Low/Undetectable
NCI-H2373	8	92	Homozygous deletion	Low/Undetectable
SDM103T2	26	82	Homozygous truncation	Low/Undetectable
NCI-H2052	<12	93	Homozygous truncation	Low/Undetectable
ACC-MESO-1	15	84	Homozygous truncation	Low/Undetectable
SPC212	60	72	Homozygous truncation	Low/Undetectable
JU77	74	82	Homozyous deleterious mut	Low/Undetectable
Mero-48a	38	109	No mutation detected	Low/Undetectable
ZL34	20	95	No mutation detected	Low/Undetectable
ZL55	48	87	No mutation detected	Low/Undetectable
Mero-14	60	94	No mutation detected	Low/Undetectable
Mero-82	113	75	No mutation detected	Low/Undetectable
ONE58	186	67	No mutation detected	Low/Undetectable
Mero-83	118	75	No mutation detected	Low/Undetectable
Mero-95	197	76	No mutation detected	Low/Undetectable
Mero-41	260	71	No mutation detected	Low/Undetectable
ZL5	178	80	Heterozygous truncation	Low/Undetectable
SPC111	1511	64	Heterozygous truncation	Low/Undetectable
NO36	>3000	71	No mutation detected	Low/Undetectable
Mero-84	>3000	33	No mutation detected	Low/Undetectable
ACC-MESO-4	879	67	No mutation detected	Detected
Mero-25	2494	54	No mutation detected	Detected
NCI-H28	>3000		No mutation detected	Detected
NCI-H2452	>3000		No mutation detected	Detected
HMMME	>3000		No mutation detected	Detected

B

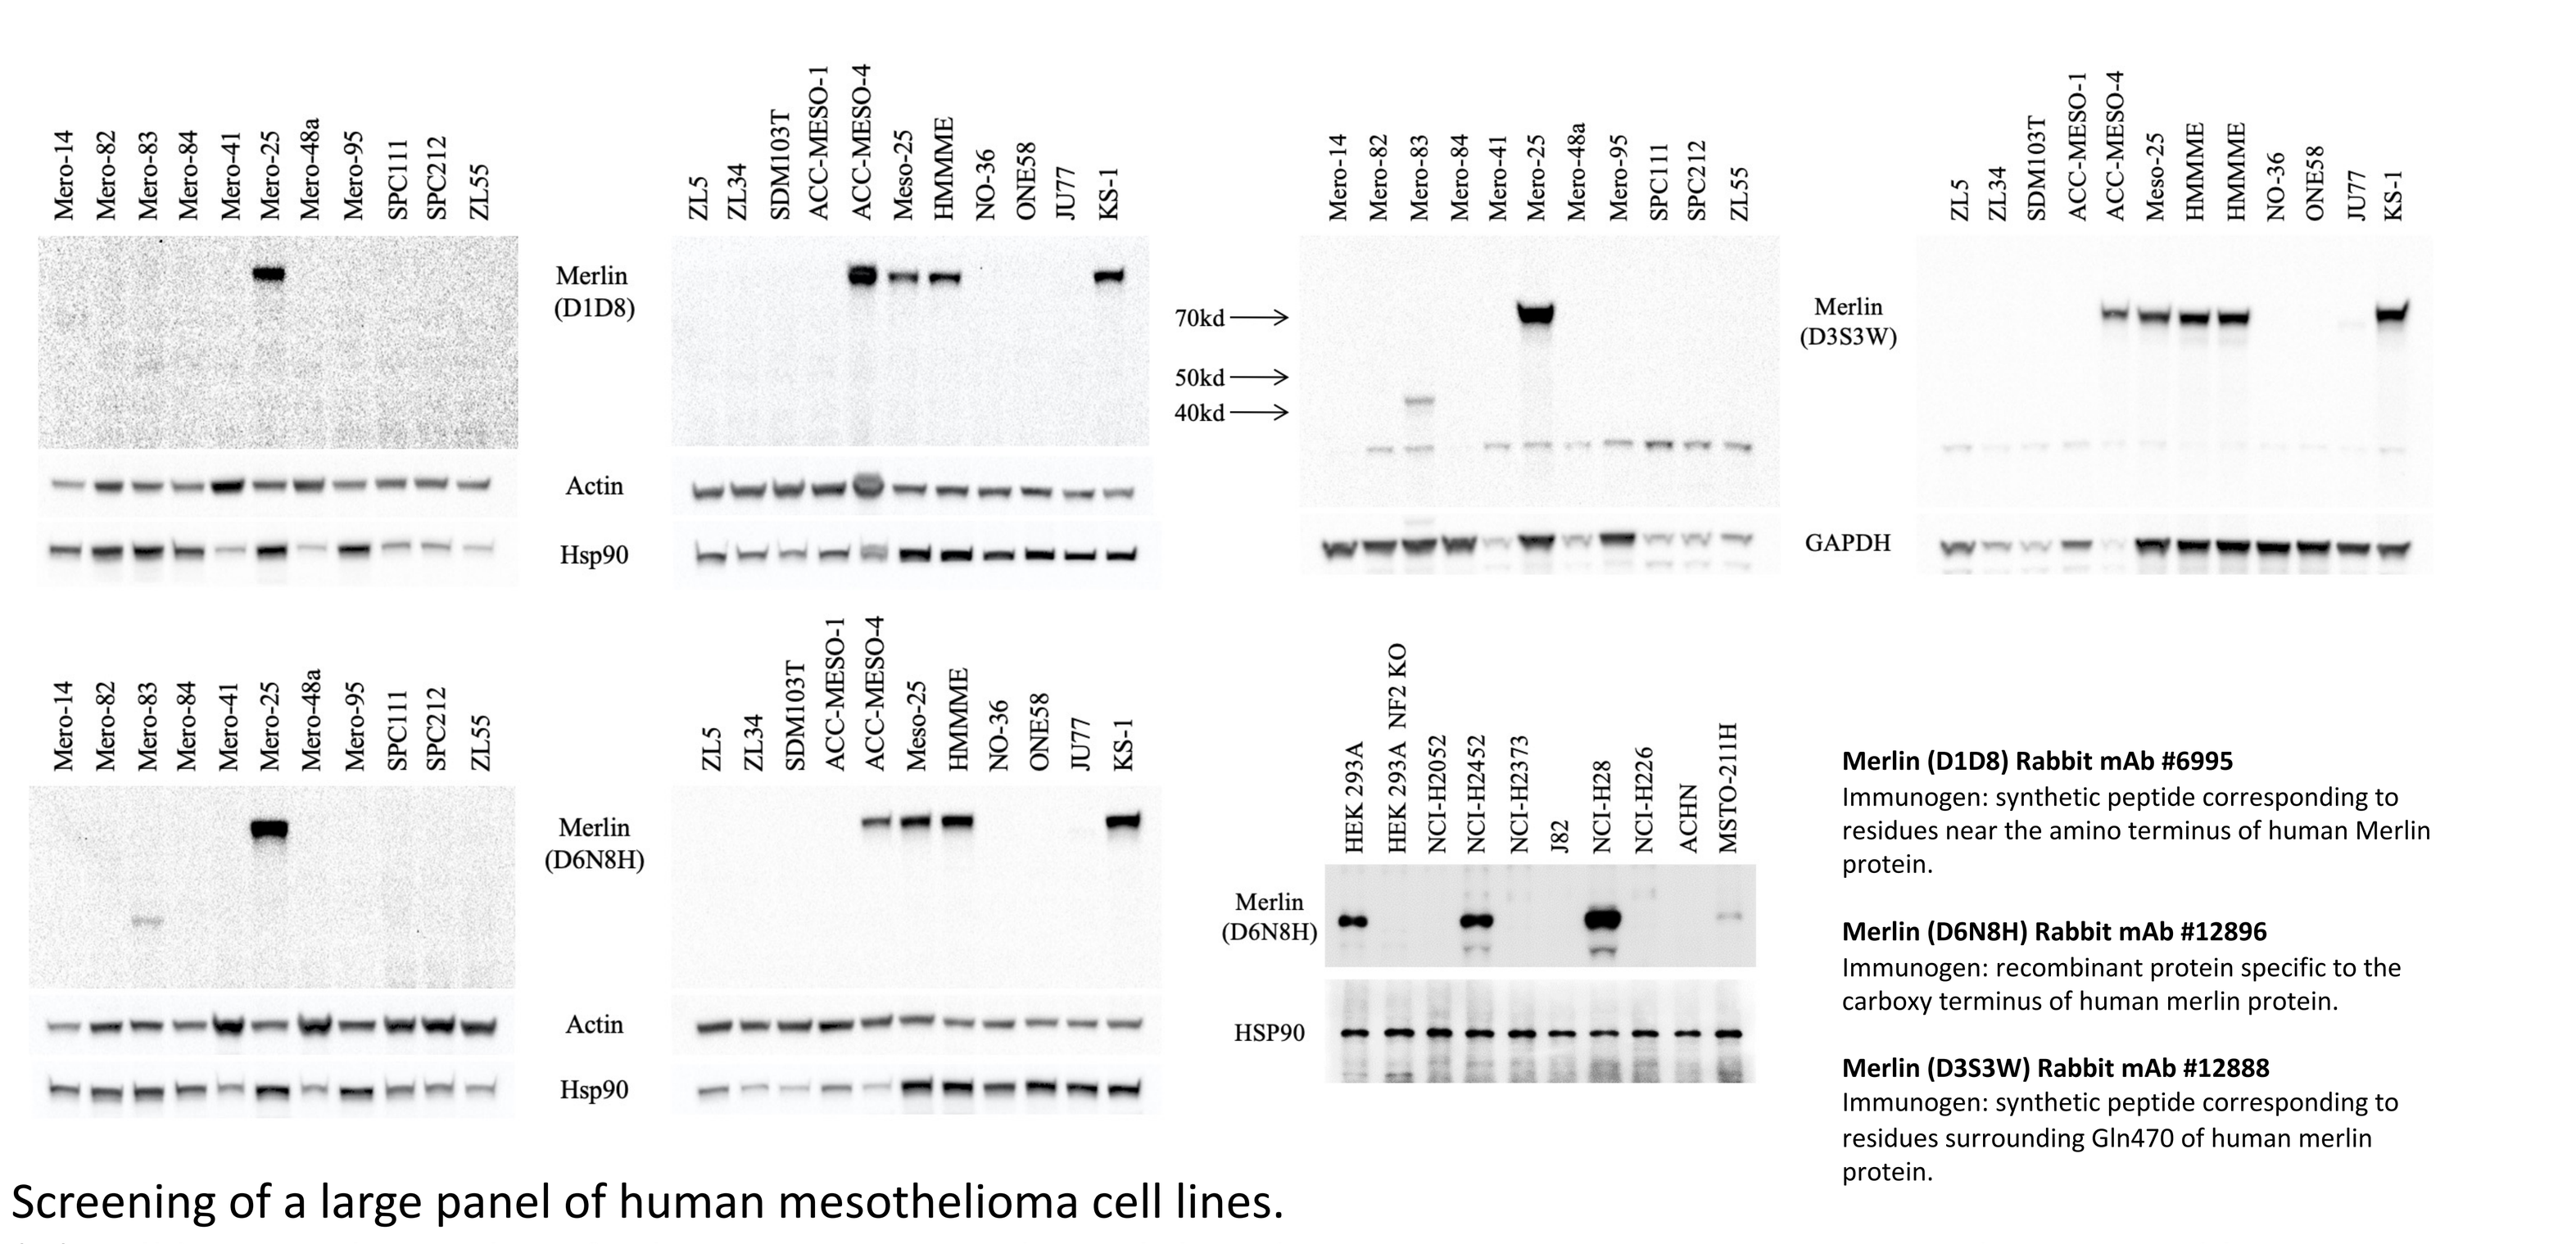

Mero-14
Mero-82
Mero-83
Mero-84
Mero-41
Mero-25
Mero-48a
Mero-95
SPC111
SPC212
ZL55
ZL5
ZL34
SDM103T
ACC-MESO-1
ACC-MESO-4
Meso-25
HMMME
NO-36
ONE58
JU77
KS-1
Merlin (D1D8)
Merlin (D6N8H)
Merlin (D3S3W)
Actin
Hsp90
GAPDH
70kd
50kd
40kd
HEK 293A
HEK 293A NF2 KO
NCI-H2052
NCI-H2452
NCI-H2373
J82
NCI-H28
NCI-H226
ACHN
MSTO-211H
HSP90

Merlin (D1D8) Rabbit mAb #6995
Immunogen: synthetic peptide corresponding to residues near the amino terminus of human Merlin protein.

Merlin (D6N8H) Rabbit mAb #12896
Immunogen: recombinant protein specific to the carboxy terminus of human merlin protein.

Merlin (D3S3W) Rabbit mAb #12888
Immunogen: synthetic peptide corresponding to residues surrounding Gln470 of human merlin protein.

Screening of a large panel of human mesothelioma cell lines.
(**A**) Cell lines without identified *NF2* mutations also exhibited *in vitro* sensitivity to VT3989 as determined by Celltiter-glo luminescent cell proliferation assay. (**B**) Western blot analysis showed that these sensitive cell lines without detectable *NF2* mutations were negative for Merlin protein expression.

VT3989 clinical responses in patients with loss of function *NF2* mutations, as well as in patients without detected *NF2* mutations based on NGS-based local laboratory testing

Initial Dose (mg/day) & Schedule	Tumor Type	*NF2* Mutation	RECIST v1.1 Response	% Change in Target Lesions	Treatment Duration (months)
200mg 2 weeks on, 2 weeks off	Dual Pleural/ Peritoneal Meso	No mutation	cPR	-81%	12+
50mg x 15 days, then once weekly	Peritoneal Meso	Unknown	cPR	-55%	6.5+
50mg continuously	Pericardial Meso	s*NF2*m	cPR	-47%	7.4
50mg continuously	Peritoneal Meso	No Mutation	cPR	-39%	7.6
100mg continuously	Peritoneal Meso	No Mutation	cPR	-39%	21+
200mg continuously	Sarcoma	sNF2m	cPR	-35%	8
150mg continuously	Peritoneal Meso	sNF2m	uPR	-30%	6.1
100mg continuously	Nasopharyngeal	sNF2m	SD	-24%	7.4
150 for 1 week on, 3 weeks off	EHE	Unknown	SD	-22%	9.5+

s*NF2*m: Somatic *NF2* mutation; cPR: confirmed partial response; uPR: unconfirmed PR; SD: stable disease

51 y/o male with advanced mesothelioma of peritoneum
Without *NF2* mutation

VT3989 50 mg PO QD; 21-day cycle

Prior therapies
- Cisplatin + Pemetrexed
- Pembrolizumab
- Carboplatin + Pemetrexed
- Pemetrexed maintenance
- Ipilimumab + Nivolumab

RECIST sustained PR (–38.7%)
On treatment for 21+ months

Patient is still on treatment C56D1 as of Sept 12, 2024

Baseline 163 mm

C22D1 100 mm

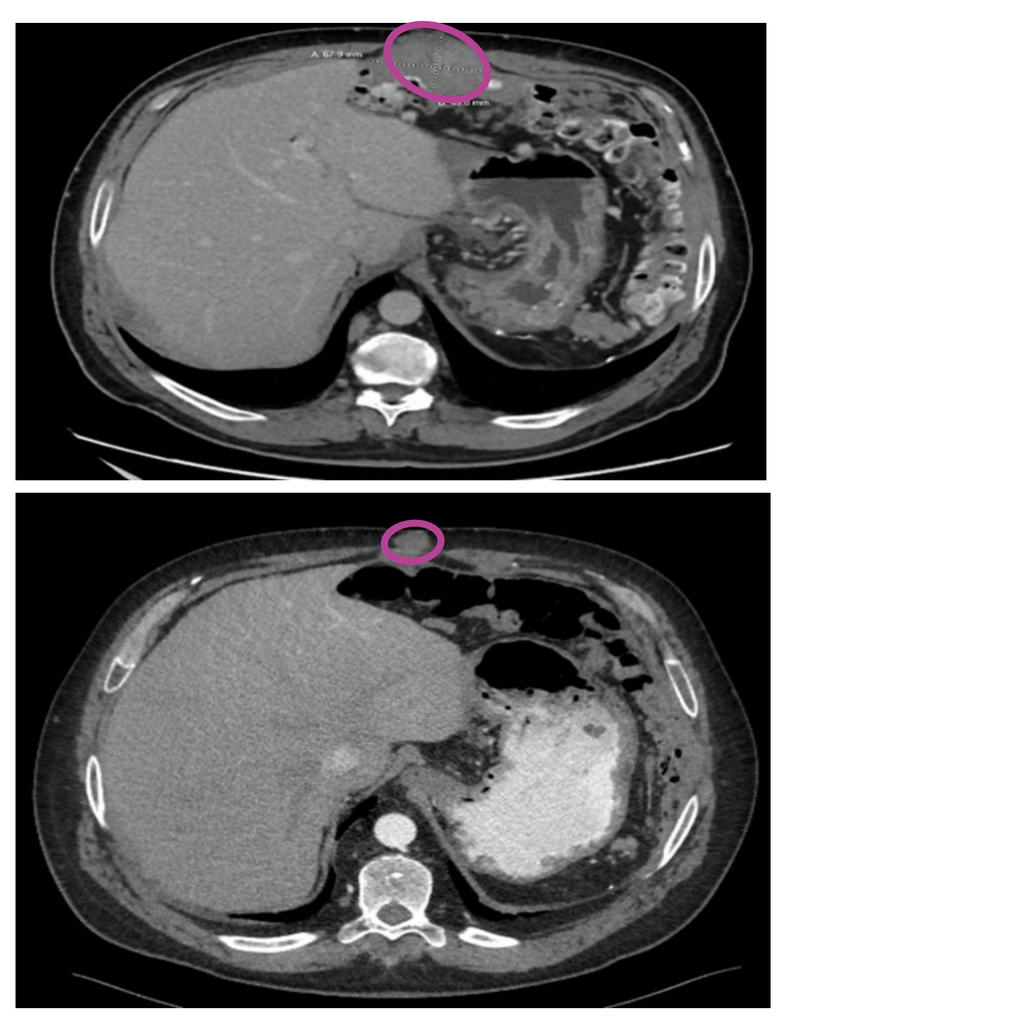

Yap *et al*, AACR 2023

VT3989 ongoing phase 1 clinical trial (NCT04665206) data were presented at 2023 AACR Annual Meeting.

Merlin-YAP Duplex IHC Assay Development and Validation

A

*Merlin and YAP **Single** Label IHC Assay Development*

- Screened multiple commercial monoclonal antibodies on the Leica BOND III IHC staining platform.
- Tested antigen-retrieval tissue pretreatments (no epitope retrieval (ER0), low pH (Citrate-based, pH 6.0-6.2) BOND Epitope Retrieval Solution 1 (ER1), and high pH (Tris-EDTA, pH 9) BOND Epitope Retrieval Solution 2 (ER2) with and without subsequent digestion with Proteinase K (PTK) enzyme) for each antibody in FFPE samples (mesothelioma cell lines, human mesothelioma and normal lung tissues).
- Tested different antibody concentrations and incubation times.

*Merlin and YAP **Dual** Label IHC Assay Development*

- Used optimized singleplex assay conditions as a starting point. In duplex assay, Leica BOND Polymer Refine Detection kit was used for detection of Merlin by DAB chromogen and Leica BOND Polymer Refine Red Detection kit for detection of YAP by Fast Red chromogen.
- Tested chosen Merlin and YAP antibodies individually within the duplex assay at various concentrations to determine optimal dilutions and demonstrate the range and linearity.
- Staining accuracy within the duplex assay was determined by use of a panel of additional human mesothelioma tissues and control cell lines.
- Duplex IHC assay development was performed under GCLP conditions in Discovery's CLIA-certified laboratory.

Merlin and YAP Dual Label IHC Assay Sensitivity and Specificity

- Tumor Sensitivity Screen of 40 mesothelioma tissue samples (40 duplex IHC tests).
- Specificity Testing on a tissue micro-array (TMA) for multi-normal human tissues.
- Each sample was reviewed by a board-certified pathologist for the range of staining intensities and patterns for each marker and scored semi-quantitatively (Percent Score and H-score). Only tumor-associated Merlin staining is scored by the pathologist; stromal staining, necrotic cells, and immune cell (lymphocyte) staining is excluded for scoring.

*Merlin and YAP Dual Label IHC Assay Analysis of Baseline **Patient Tumor Samples** from **VT3989 Clinical Trial***

B

NCI-H28: High Merlin | NCI-H2452: Moderate Merlin | NCI-H2052: Low/neg Merlin

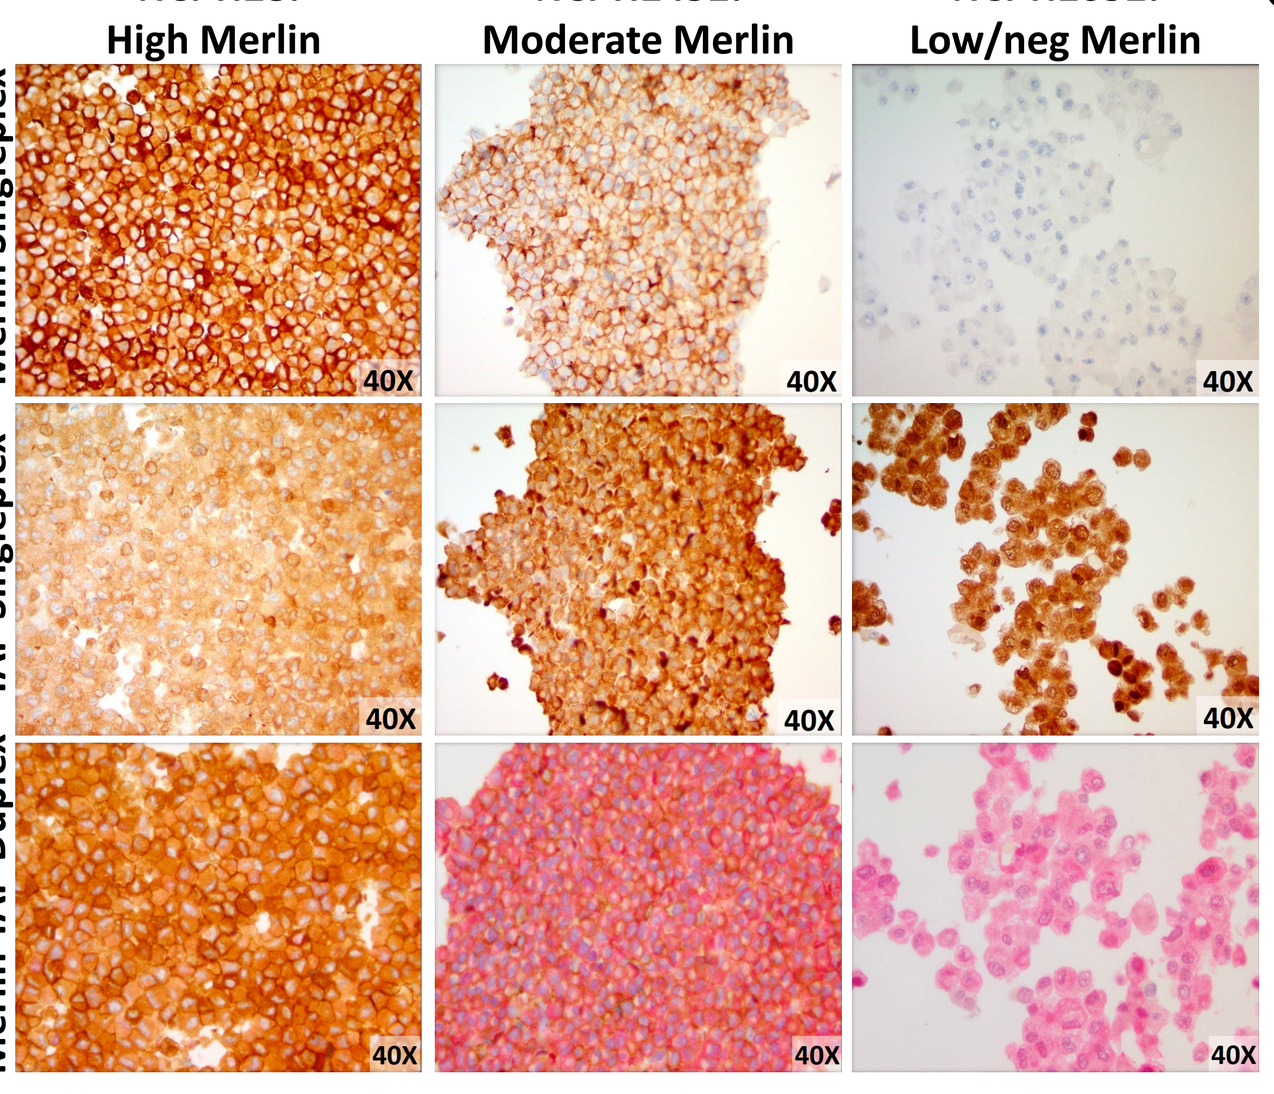

Merlin Singleplex
YAP Singleplex
Merlin-YAP Duplex
40X

C

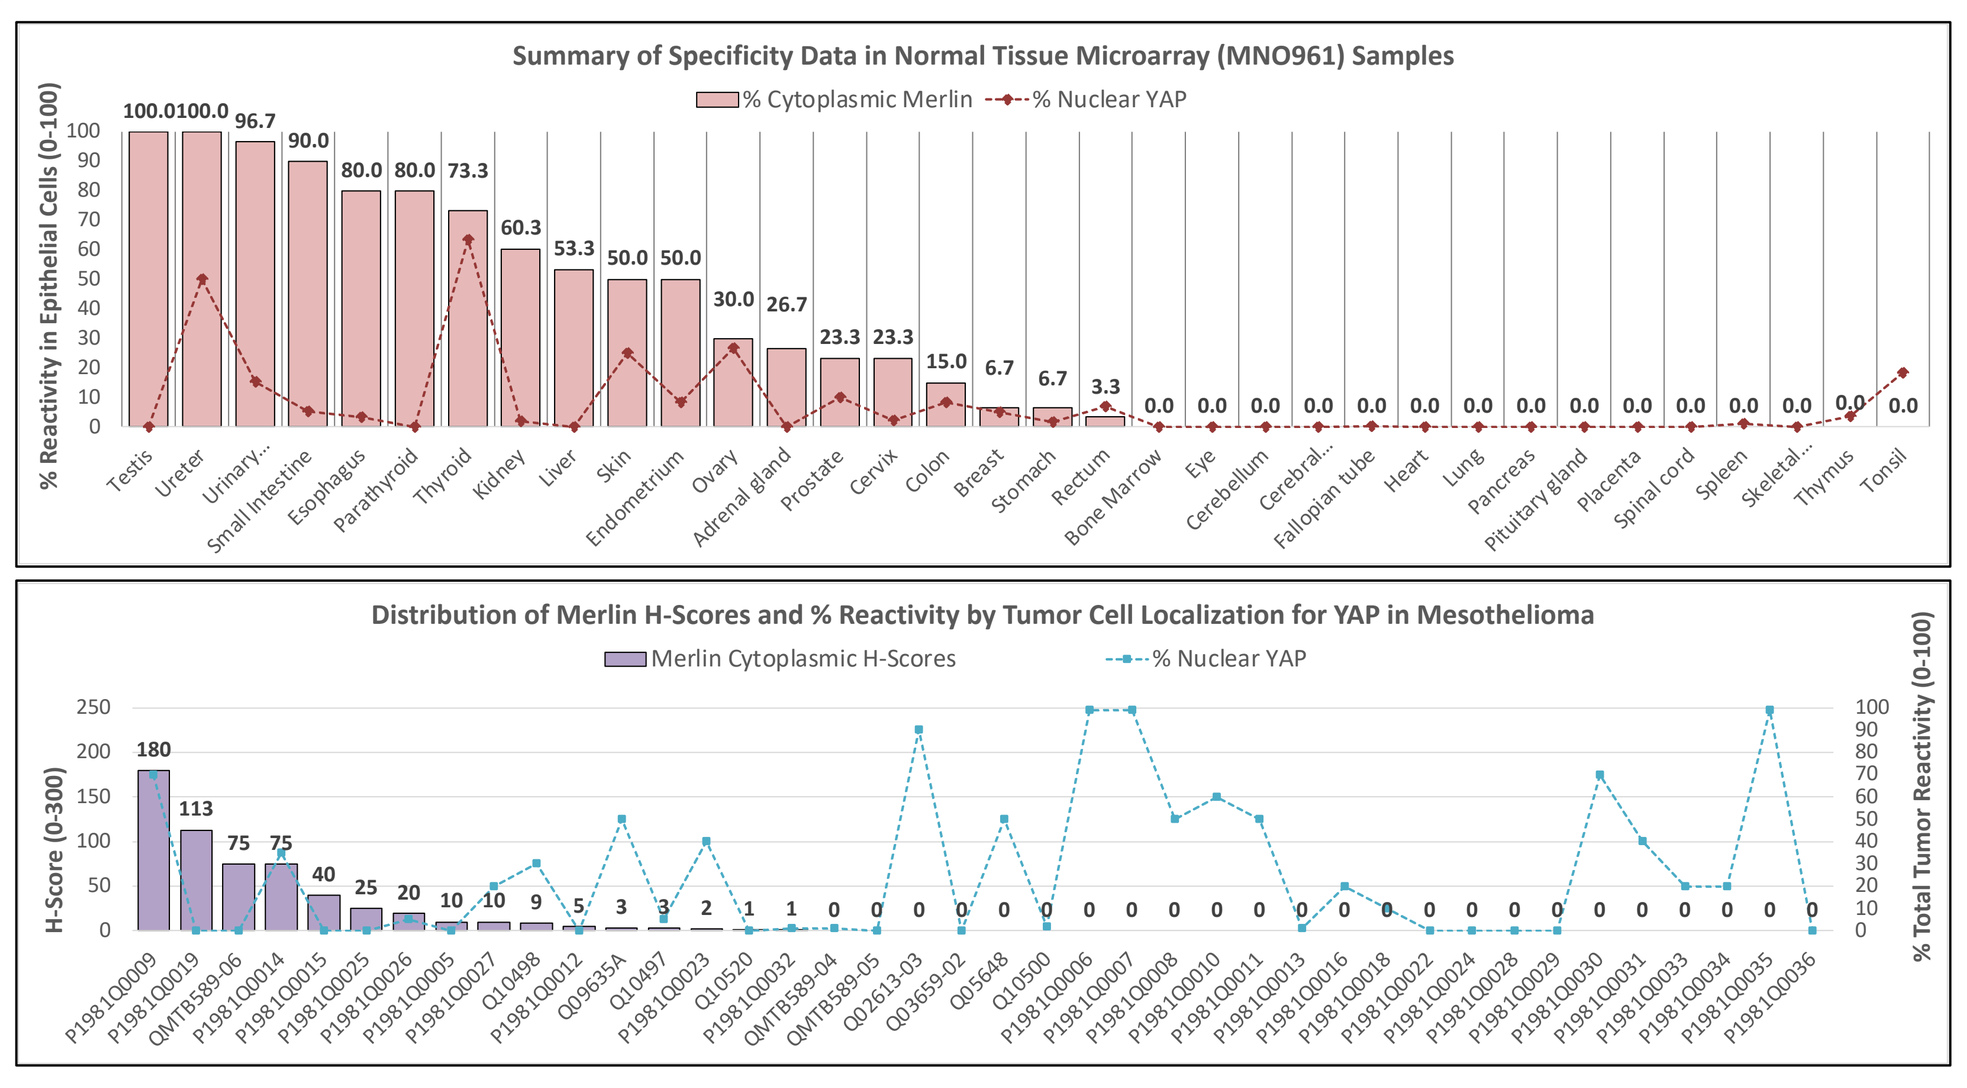

Summary of Specificity Data in Normal Tissue Microarray (MNO961) Samples
% Cytoplasmic Merlin
% Nuclear YAP
% Reactivity in Epithelial Cells (0-100)
Distribution of Merlin H-Scores and % Reactivity by Tumor Cell Localization for YAP in Mesothelioma
Merlin Cytoplasmic H-Scores
H-Score (0-300)
% Total Tumor Reactivity (0-100)

D

P1981Q0006: NF2_p.E89fs Frameshift - LOF | P1981Q0010: No NF2 mutation detected | P1981Q0015: No NF2 mutation detected | P1981Q0019: No NF2 mutation detected

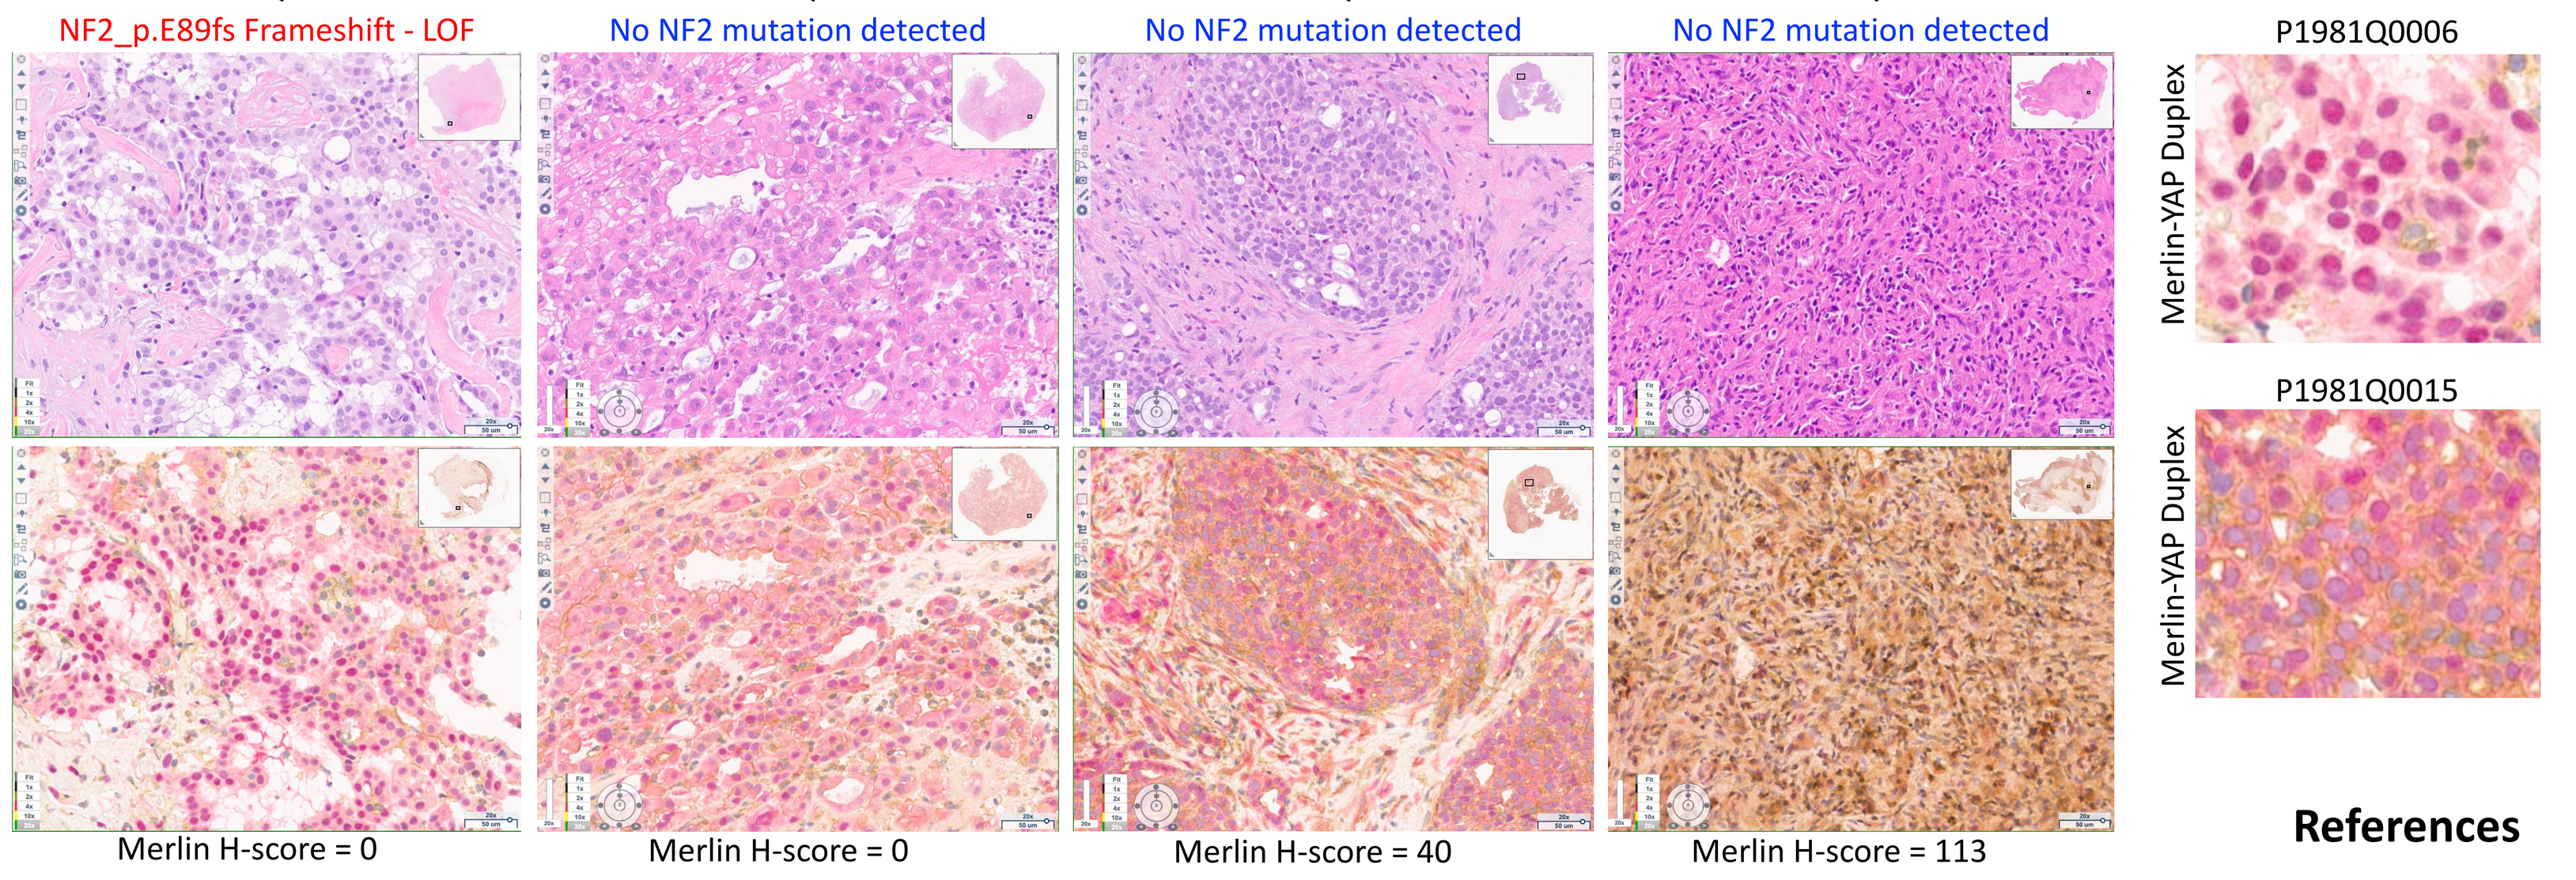

H&E
Merlin (DAB); YAP (Red)
P1981Q0006
P1981Q0015
Merlin-YAP Duplex

Merlin H-score = 0 | Merlin H-score = 0 | Merlin H-score = 40 | Merlin H-score = 113

Merlin IHC assays show that majority of mesothelioma patients with no detected *NF2* mutations have low or no Merlin expression

Three VT3989 partial response (PR) mesotheliomas without detected NF2 mutations have negative or low Merlin expression.

VT3989 Clinical Trial (Subset of Patients)	Best Response	% Change Target Lesion	% Cytoplasmic YAP and Merlin (in *Duplex* assay)	% Cytoplasmic Merlin Only (in *Singleplex* assay)	Merlin H-score from *Merlin-YAP Duplex* IHC	Merlin H-score from *Merlin Singleplex* IHC
Patients with NF2 mutation N=4	PR	-48.4%	75		5	not done
	PR	-30%	0	0	0	0
	SD	0.9%	50		60	not done
	PD	35.5%	5		0	not done
Patients with no NF2 mutation detected N=18	**PR**	**-70.7%**	**5**		**60**	not done
	PR	**-49%**	**100**	**95**	**0**	**100***
	PR	**-31.3%**	**10**		**0**	not done
	SD	-12.8%	N/A		N/A	not done
	SD	-14.4%	20	50	100	50
	SD	-26%	70		40	not done
	SD	-13.3%	70	98	20	153
	SD	-20%	55		20	not done
	SD	16.7%	100	20	0	20
	SD	15.2%	10		0	not done
	PD	36%	20	95	135	185
	PD	31.3%	60	95	20	115
	PD	-3.2%	70		10	not done
	PD	22.1%	87	80	4	110
	PD	-13.6%	98	60	2	65
	PD	11.5%	9		1	not done
	PD	50%	100	90	0	114
	PD	71%	0		0	not done

* The cytoplasmic Merlin staining was masked by the cytoplasmic YAP staining in the duplex IHC assay and was excluded from scoring; hence, Merlin H-score was 0. In singleplex IHC assay, Merlin H-score was 100 (95% Staining ≥1+; 5% Staining ≥2+; 0% Staining ≥3+) with low intensity. Yellow highlights Merin H-score >100 (Duplex: 1/18; Singleplex: 5/18).

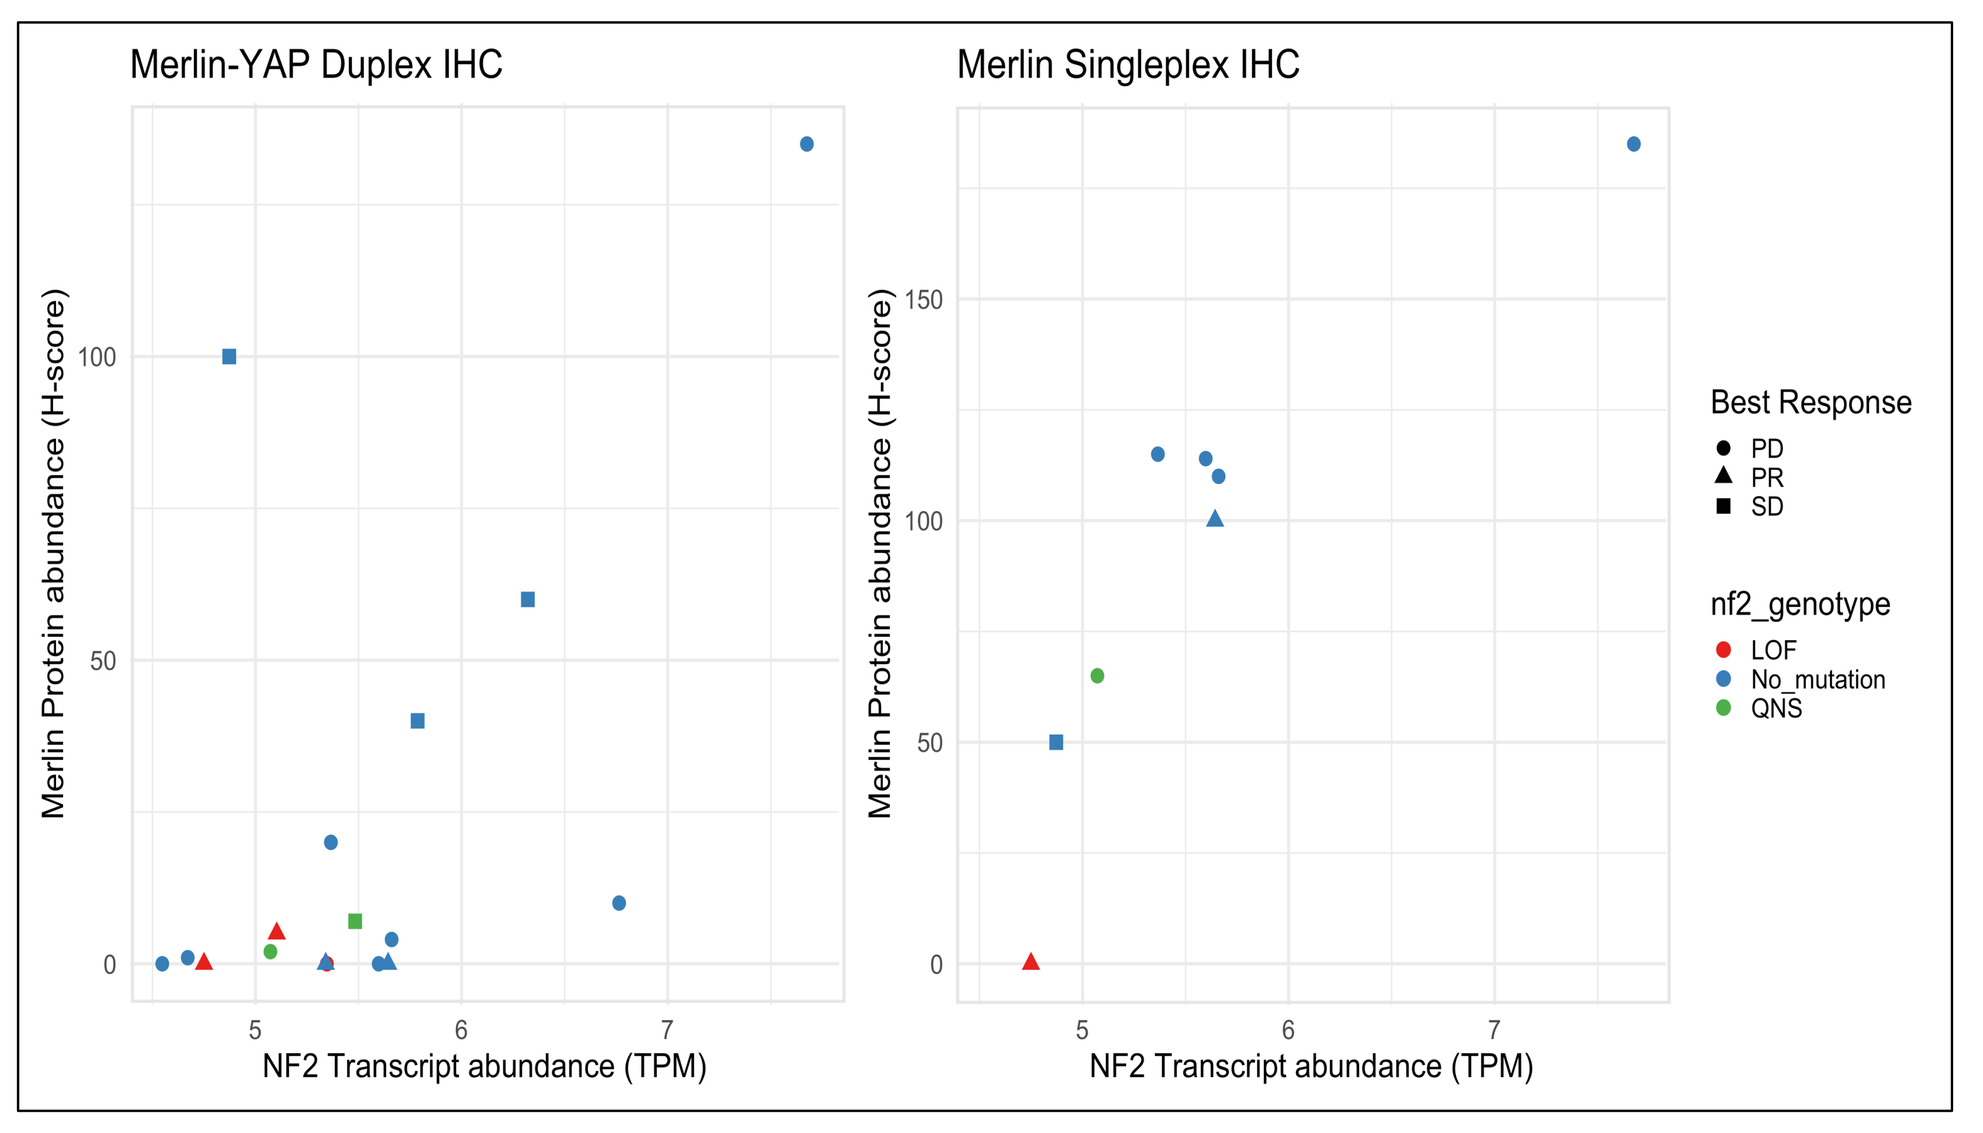

Merlin-YAP Duplex IHC
Merlin Singleplex IHC
Merlin Protein abundance (H-score)
NF2 Transcript abundance (TPM)
Best Response
PD
PR
SD
nf2_genotype
LOF
No_mutation
QNS

Transcriptome expression was evaluated in parallel using a RNASeq platform, and RNA and protein expression were compared. Each data point in the plots represents an individual patient. Only a subset of trial patients with materials available for all assays (IHC NGS, and RNAseq) were included in this analysis. Best Response=Best response to VT3989. PD=Progressive Disease. PR=Partial Response. SD=Stable Disease.

Case 1: Partial Response NF2_p.E34fs Frameshift – LOF | **Case 2: Partial Response** No NF2 mutation detected | **Case 3: Progressive Disease** No NF2 mutation detected | **Case 4: Progressive Disease** No NF2 mutation detected

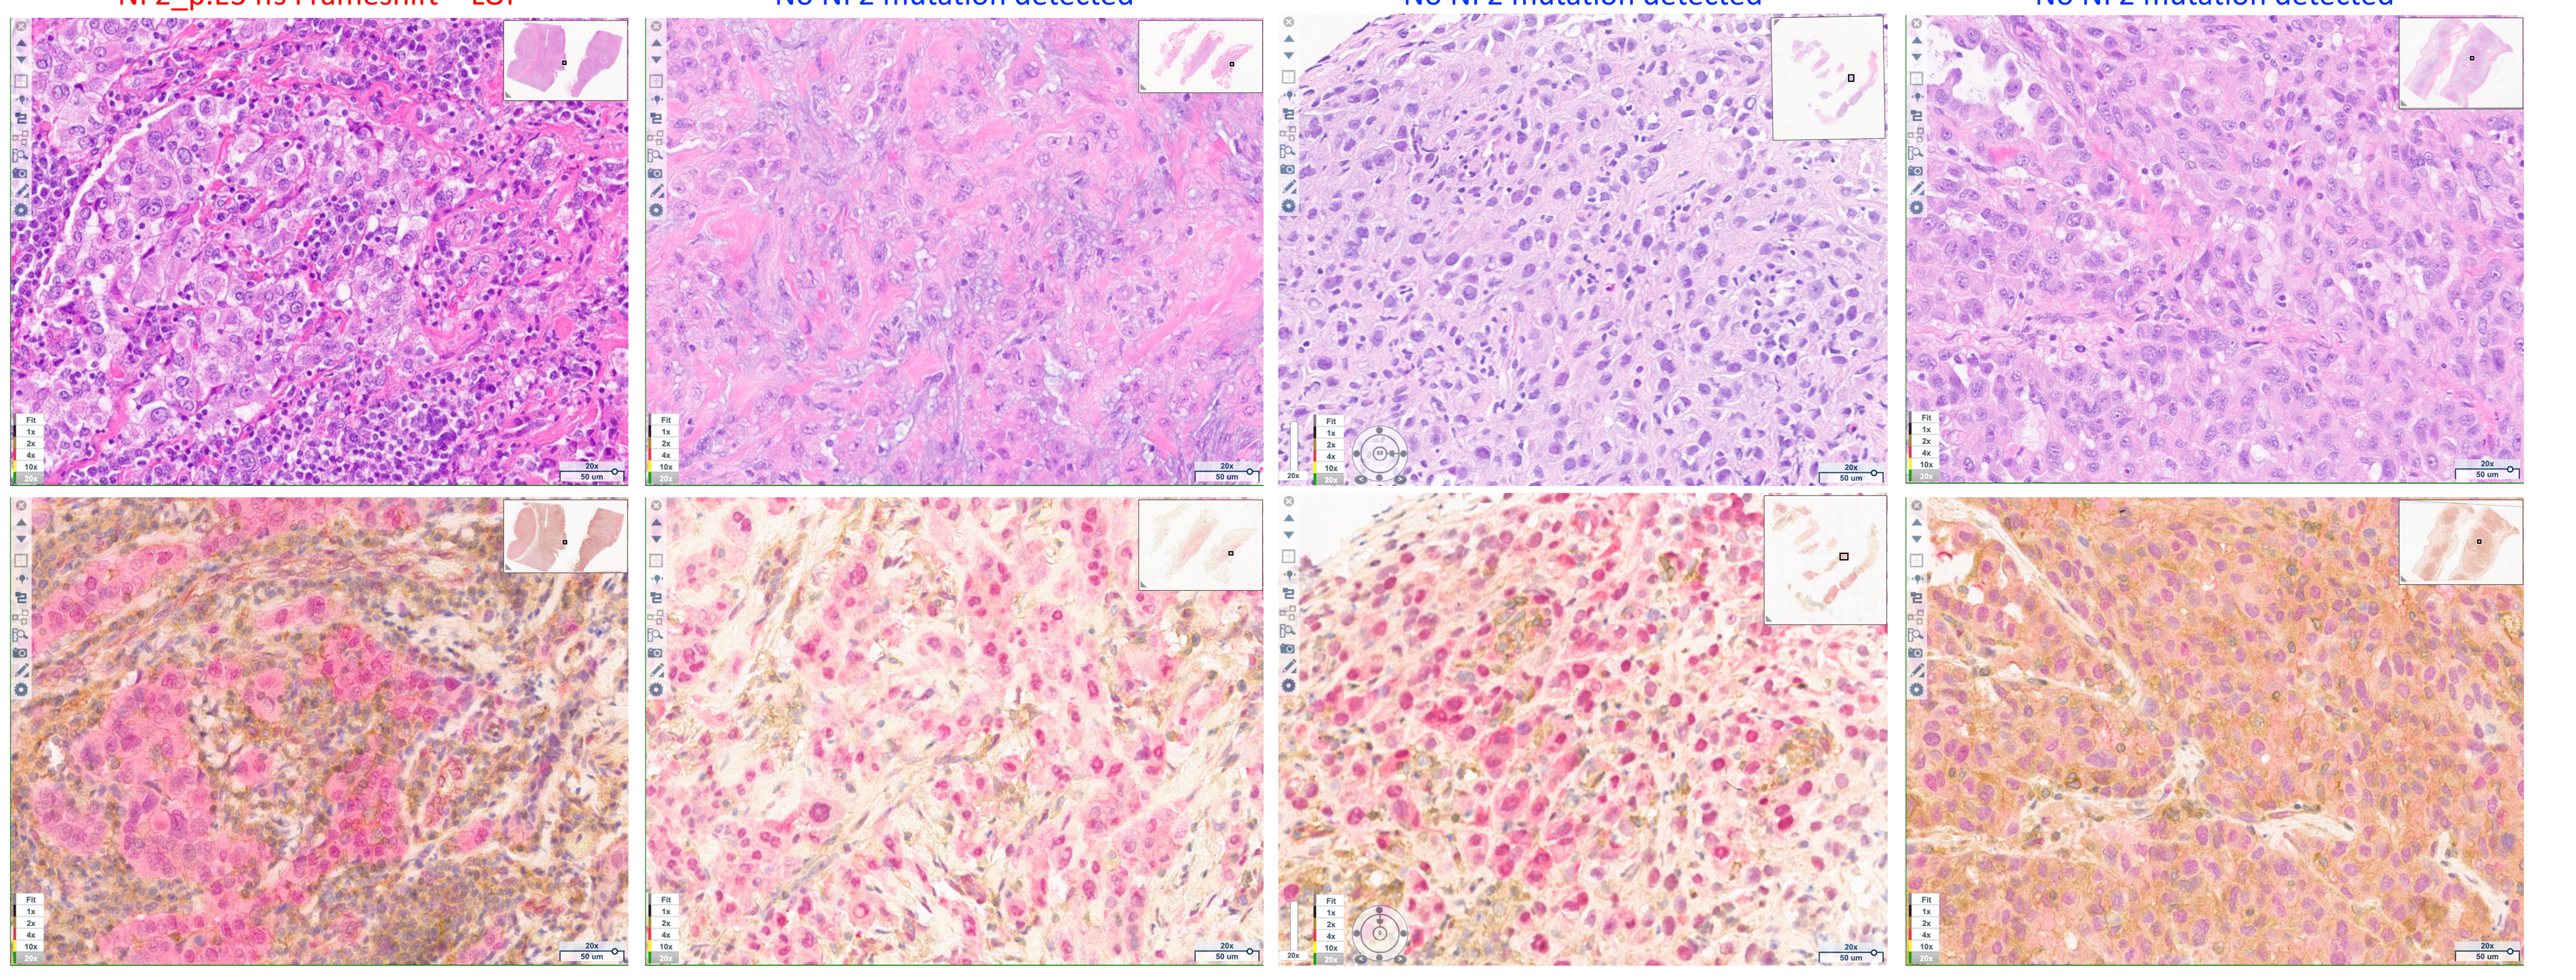

Case 1: Merlin H-Score = 0. Lymphocyte staining throughout tissues not included in H-score calculation. **Case 2**: Merlin H-score = 0. No detected NF2 mutation. **Case 3**: Merlin H-score = 0. No detected NF2 mutation. **Case 4**: Merlin H-score = 0 (duplex). Merlin H-score = 114 (singleplex; 90% Staining ≥1+; 22% Staining ≥2+; 2% Staining ≥3+). Many lymphocytes staining throughout tumor.

References
1. Huh et al, 2019, Cells. 8(6):600
2. Franklin et al, 2023, Nat Rev Cancer. 23(8):512-525
3. Tang et al, 2021, Mol Cancer Ther. 20(6):986-998.
4. Li et al, 2023, Mod Pathol. 36(3):100030

36th EORTC-NCI-AACR SYMPOSIUM
Barcelona, Spain, 23-25 October 2024