VT3989, a clinical TEAD palmitoylation inhibitor, enhances the efficacy and durability of multiple targeted therapies of the MAPK and PI3K/AKT/mTOR pathways

Tracy T. Tang and Leonard Post, Vivace Therapeutics, San Mateo, California, USA

Poster #B088

Background

- TEAD transcription factors are the major effectors of the Hippo-YAP/TAZ-TEAD pathway involved in the regulation of cell proliferation, survival, and cell migration.
- TEAD auto-palmitoylation has been shown to be required for TEAD interaction with coactivator YAP/TAZ and hence activation of transcriptional activity.
- There are multiple reports that YAP/TEAD activation provides the essential survival signal in drug-tolerant persister/dormant cells in cancer in response to treatment with targeted therapies, leading to drug resistance and cancer relapse in *BRAF*-mutant, *KRAS*-mutant, *EGFR*-mutant, and *ALK*-rearranged non–small cell lung cancers (NSCLC).
- VT3989, with similar biological activity *in vitro* and *in vivo* as the potent small molecule TEAD auto-palmitoylation inhibitors reported (Tang *et al*, 2021, *Mol Cancer Ther*. 20(6):986-998), disrupts YAP/TAZ-TEAD protein interaction, suppresses TEAD transcriptional activity, and selectively blocks proliferation of *NF2*-deficient mesothelioma *in vitro* and inhibit *NF2* mutant xenograft tumor growth *in vivo*.
- VT3989, currently in clinical testing, was evaluated for effect on emerging drug tolerant persister/dominant cancer cells and drug resistance.

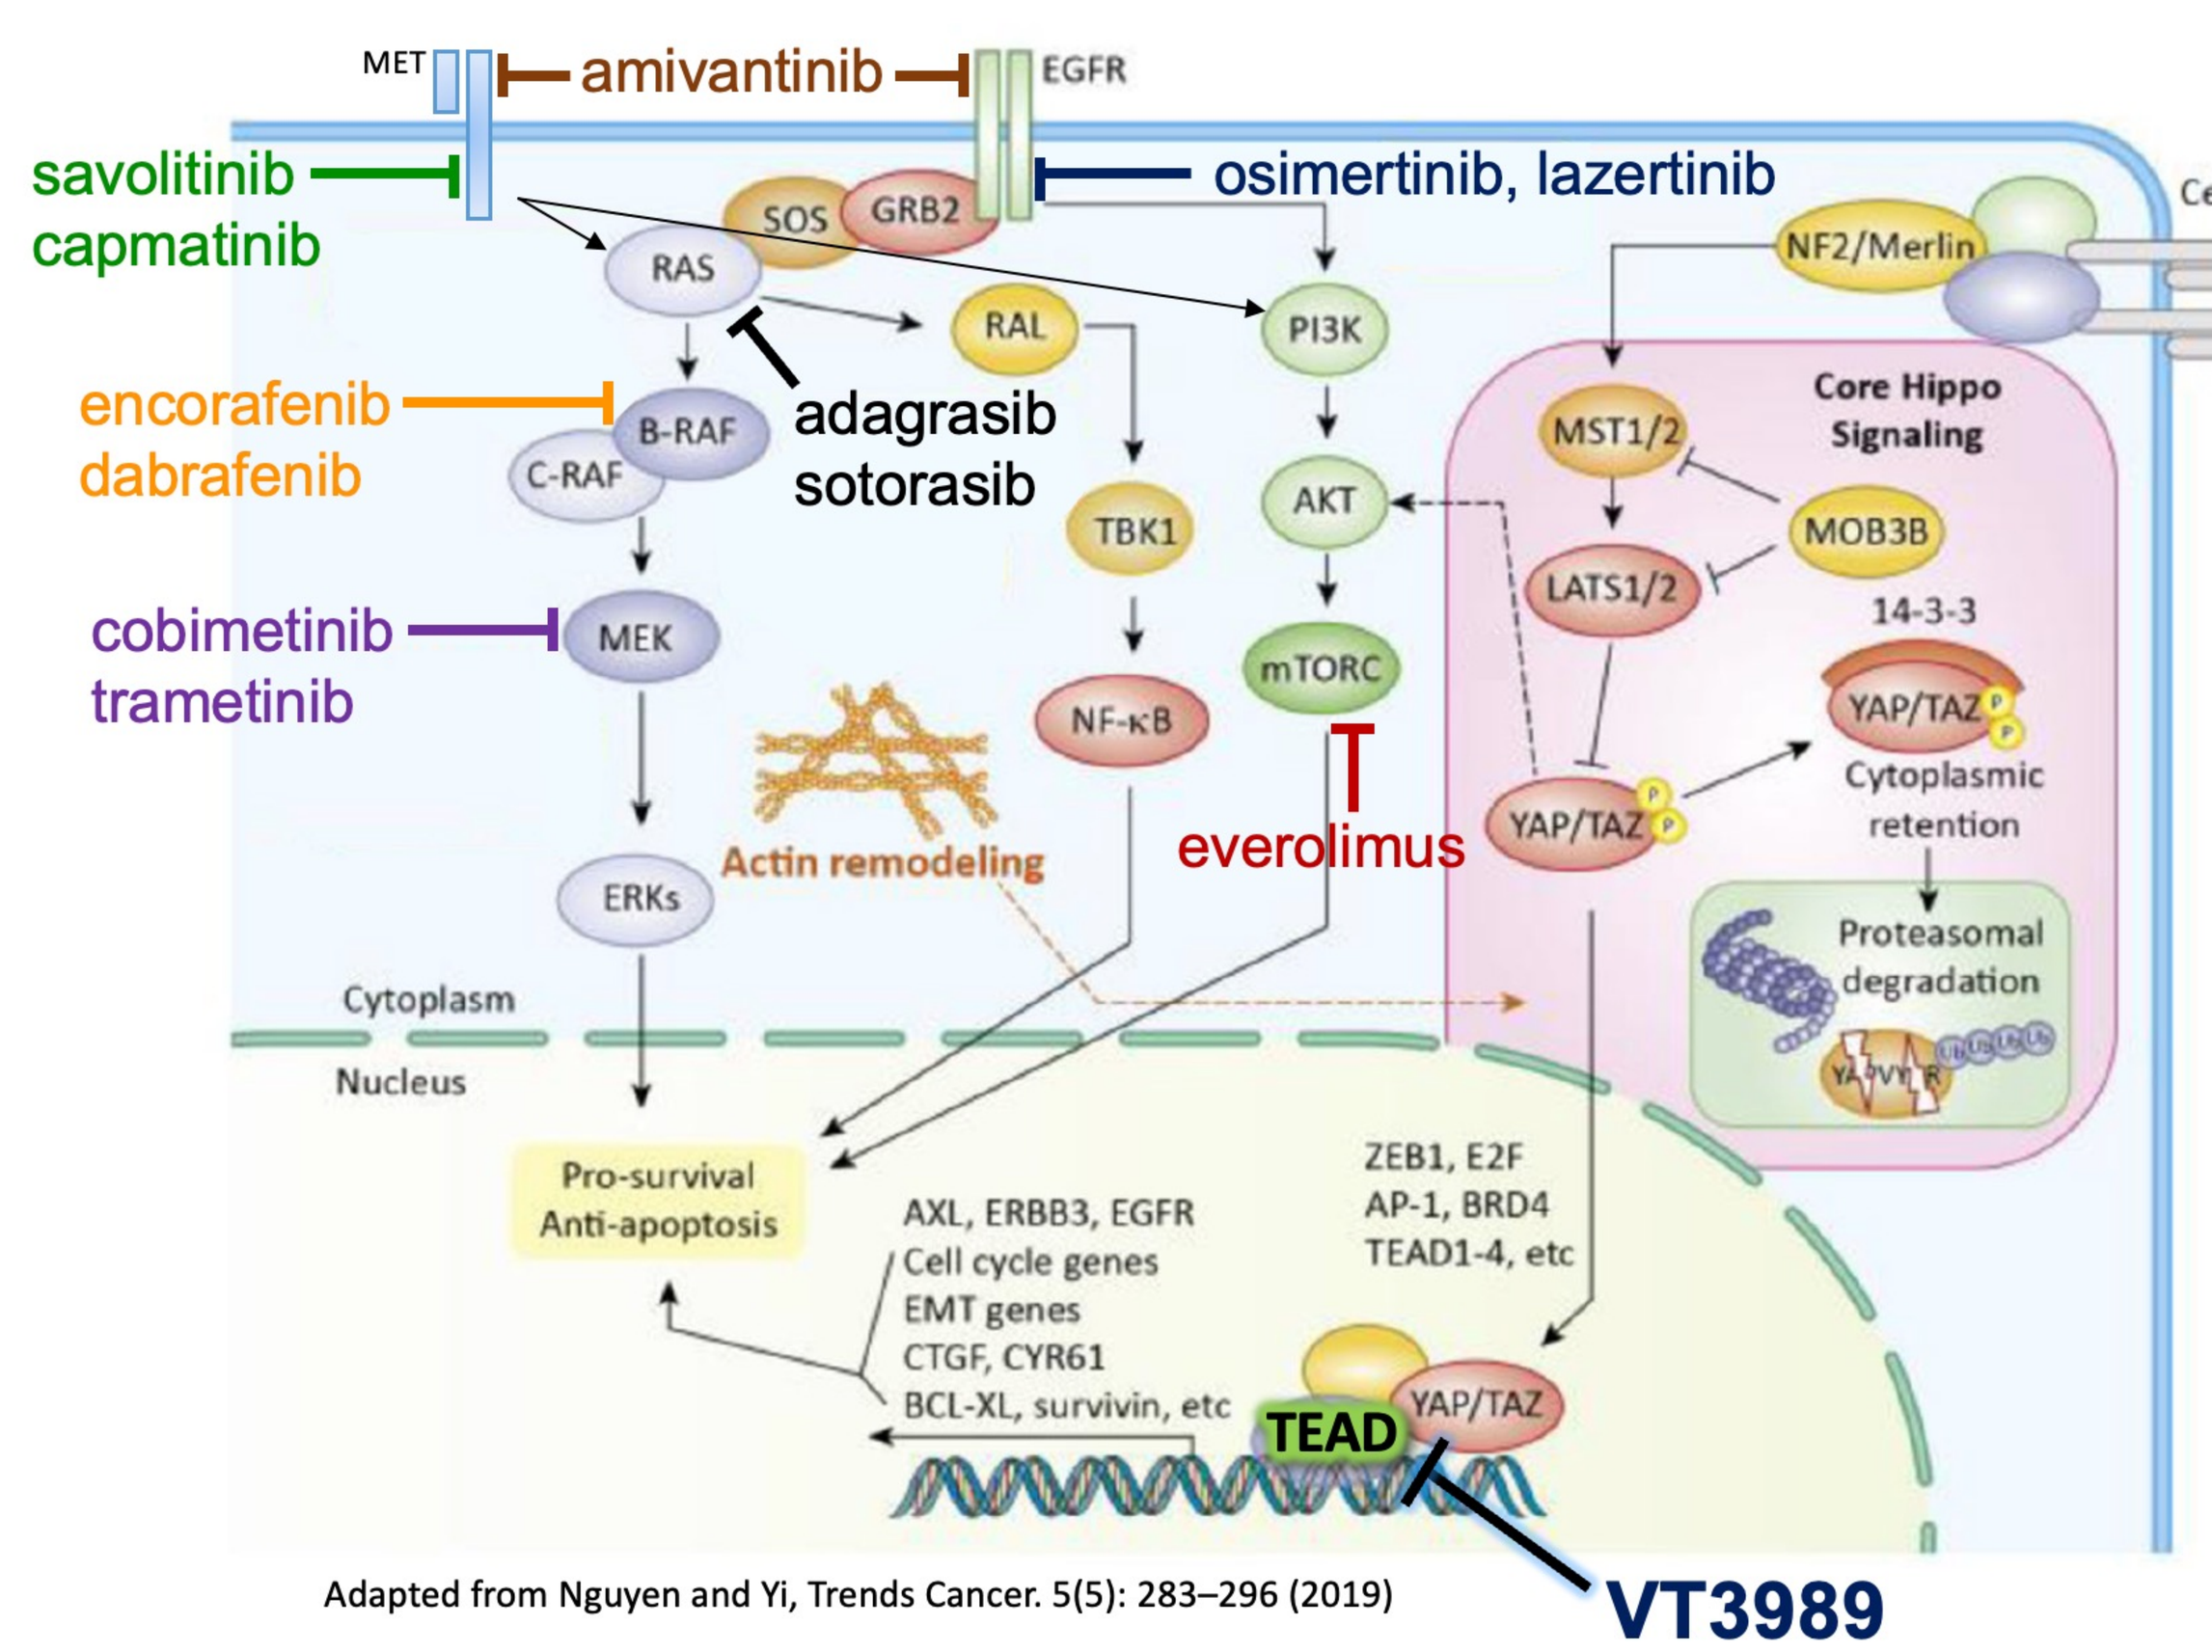

MET
amivantinib
EGFR
savolitinib
capmatinib
osimertinib, lazertinib
SOS
GRB2
RAS
RAL
PI3K
NF2/Merlin
Cell junction
encorafenib
dabrafenib
B-RAF
C-RAF
adagrasib
sotorasib
TBK1
AKT
Core Hippo Signaling
MST1/2
MOB3B
LATS1/2
14-3-3
cobimetinib
trametinib
MEK
NF-κB
mTORC
everolimus
YAP/TAZ
Cytoplasmic retention
Proteasomal degradation
Actin remodeling
ERKs
Cytoplasm
Nucleus
Pro-survival
Anti-apoptosis
AXL, ERBB3, EGFR
Cell cycle genes
EMT genes
CTGF, CYR61
BCL-XL, survivin, etc
ZEB1, E2F
AP-1, BRD4
TEAD1-4, etc
TEAD
VT3989
Vivace TEAD Inhibitors

Adapted from Nguyen and Yi, Trends Cancer. 5(5): 283–296 (2019)

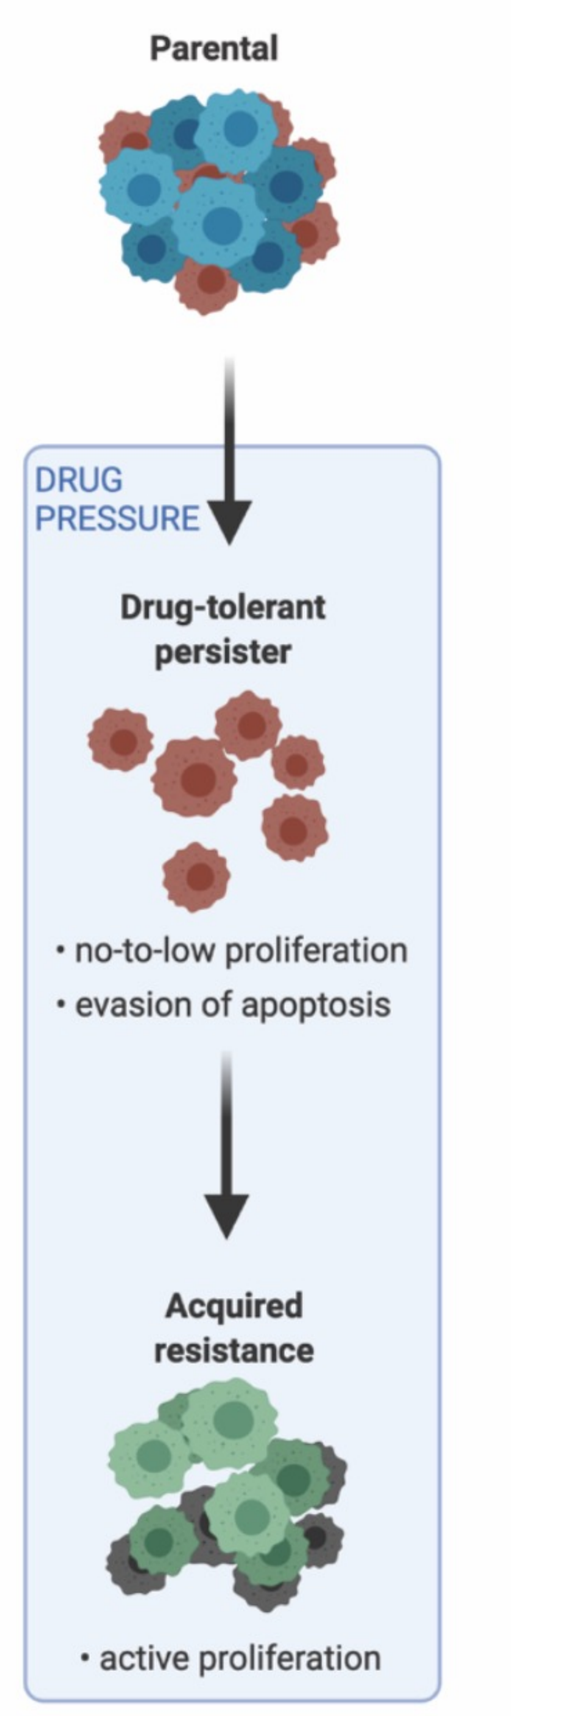

Parental
DRUG PRESSURE
Drug-tolerant persister
• no-to-low proliferation
• evasion of apoptosis
Acquired resistance
• active proliferation

Franziska Haderk and Trever Bivona Bioarxiv2021

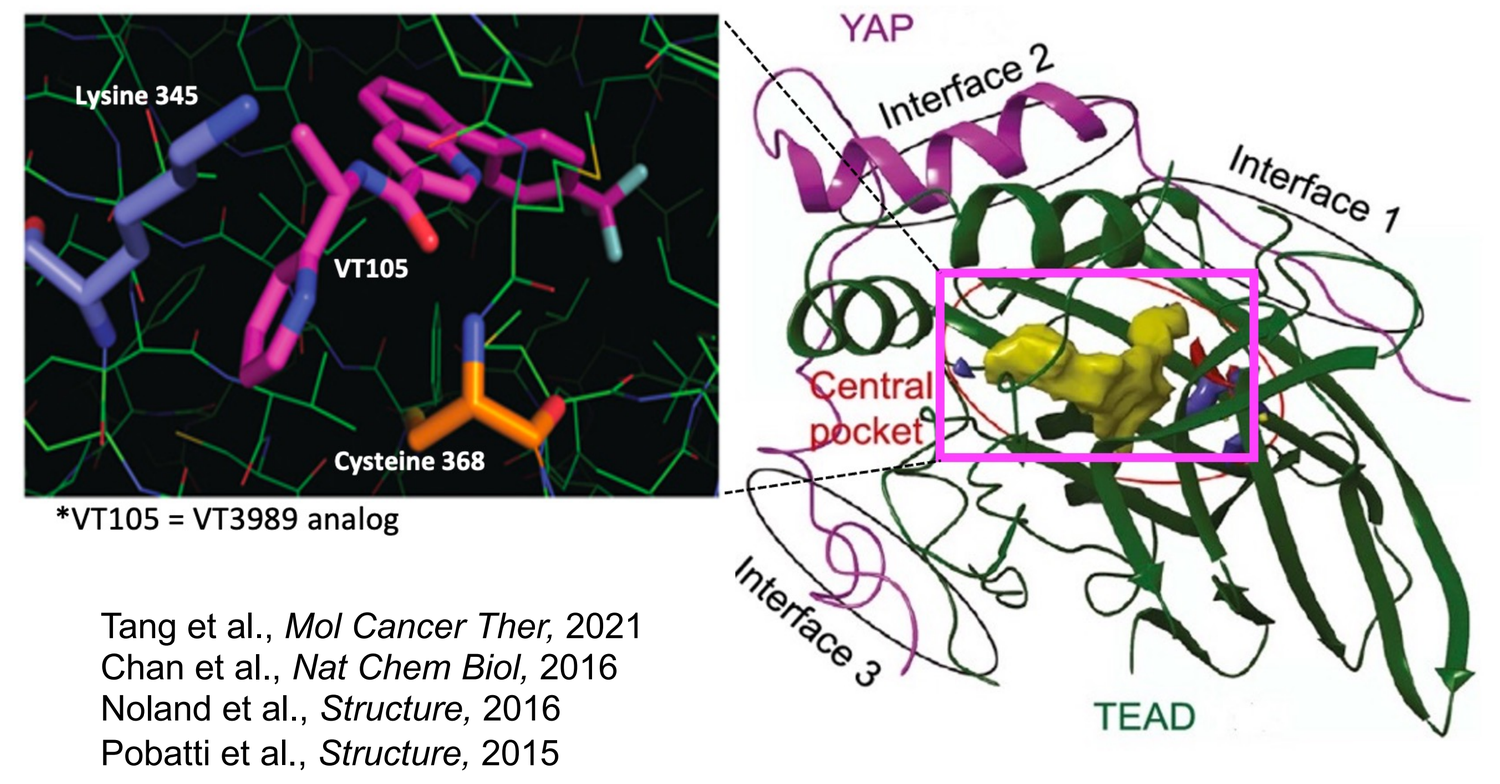

Lysine 345
VT105
Cysteine 368
YAP
Interface 2
Interface 1
Central pocket
Interface 3
TEAD

*VT105 = VT3989 analog

Tang et al., *Mol Cancer Ther*, 2021
Chan et al., *Nat Chem Biol*, 2016
Noland et al., *Structure*, 2016
Pobatti et al., *Structure*, 2015

Compound ID	VT3989	Inactive Analog
YAP IC_{50} (nM)	4.9	>3uM
TEAD1 ΔTm (°C)	11.6	0.2
TEAD2 ΔTm (°C)	8.8	0.5
TEAD3 ΔTm (°C)	8.5	0.2
TEAD4 ΔTm (°C)	4.6	0.2

YAP IC_{50} = YAP Reporter Assay IC_{50}

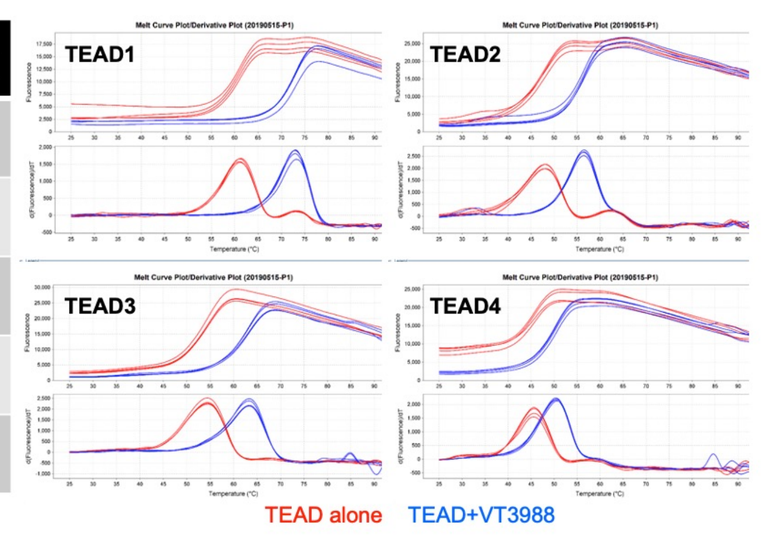

TEAD1
TEAD2
TEAD3
TEAD4
TEAD alone
TEAD+VT3988

- Palmitoylation of a conserved cysteine is required for TEAD interaction with YAP. The palmitate is buried in a central pocket in the YAP-binding domain of TEAD.

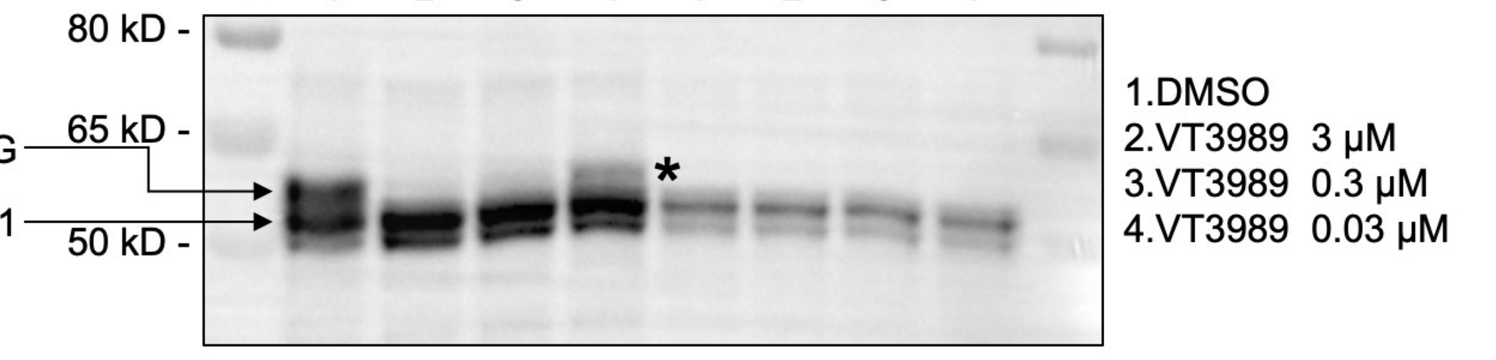

APEGS
Input
M 1 2 3 4 1 2 3 4 M
80 kD
65 kD
50 kD
TEAD1-PEG
TEAD1
*
1.DMSO
2.VT3989 3 μM
3.VT3989 0.3 μM
4.VT3989 0.03 μM

➢ **VT3989 and other Vivace TEAD inhibitors occupy the palmitate pocket in TEAD, inhibiting auto-palmitoylation and thereby inhibiting transcription function of YAP-TEAD.**

Summary and Conclusions

- **VT3989 enhances the efficacy and durability of response to clinically approved EGFR inhibitors (osimertinib, lazertinib), MET inhibitors (savolitinib, capmatinib), EGFR-MET bispecific antibody (amivantamab), KRAS-G12C inhibitors (sotorasib, adagrasib), BRAF inhibitors (encorafenib, dabrafenib), MEK inhibitors (trametinib, cobimetinib), and mTOR inhibitor (everolimus).**
- **With the addition of VT3989, combination anti-tumor effect was observed and/or tumor regrowth was delayed compared to targeted therapies alone. The persister cell mechanism seems to work in a very broad range of combinations, and the broad range of combination agents suggests a fundamental role for YAP-TEAD in the development of resistance to targeted therapy.**
- **In addition to Hippo-YAP/TAZ-TEAD pathway driven cancers, such as mesothelioma, where VT3989 can show monotherapy efficacy, future VT3989 clinical studies will include combination therapy with approved targeted therapies to enhance the efficacy and durability of treatment.**

Combination with EGFR small molecule inhibitors in EGFR mutant NSCLC models

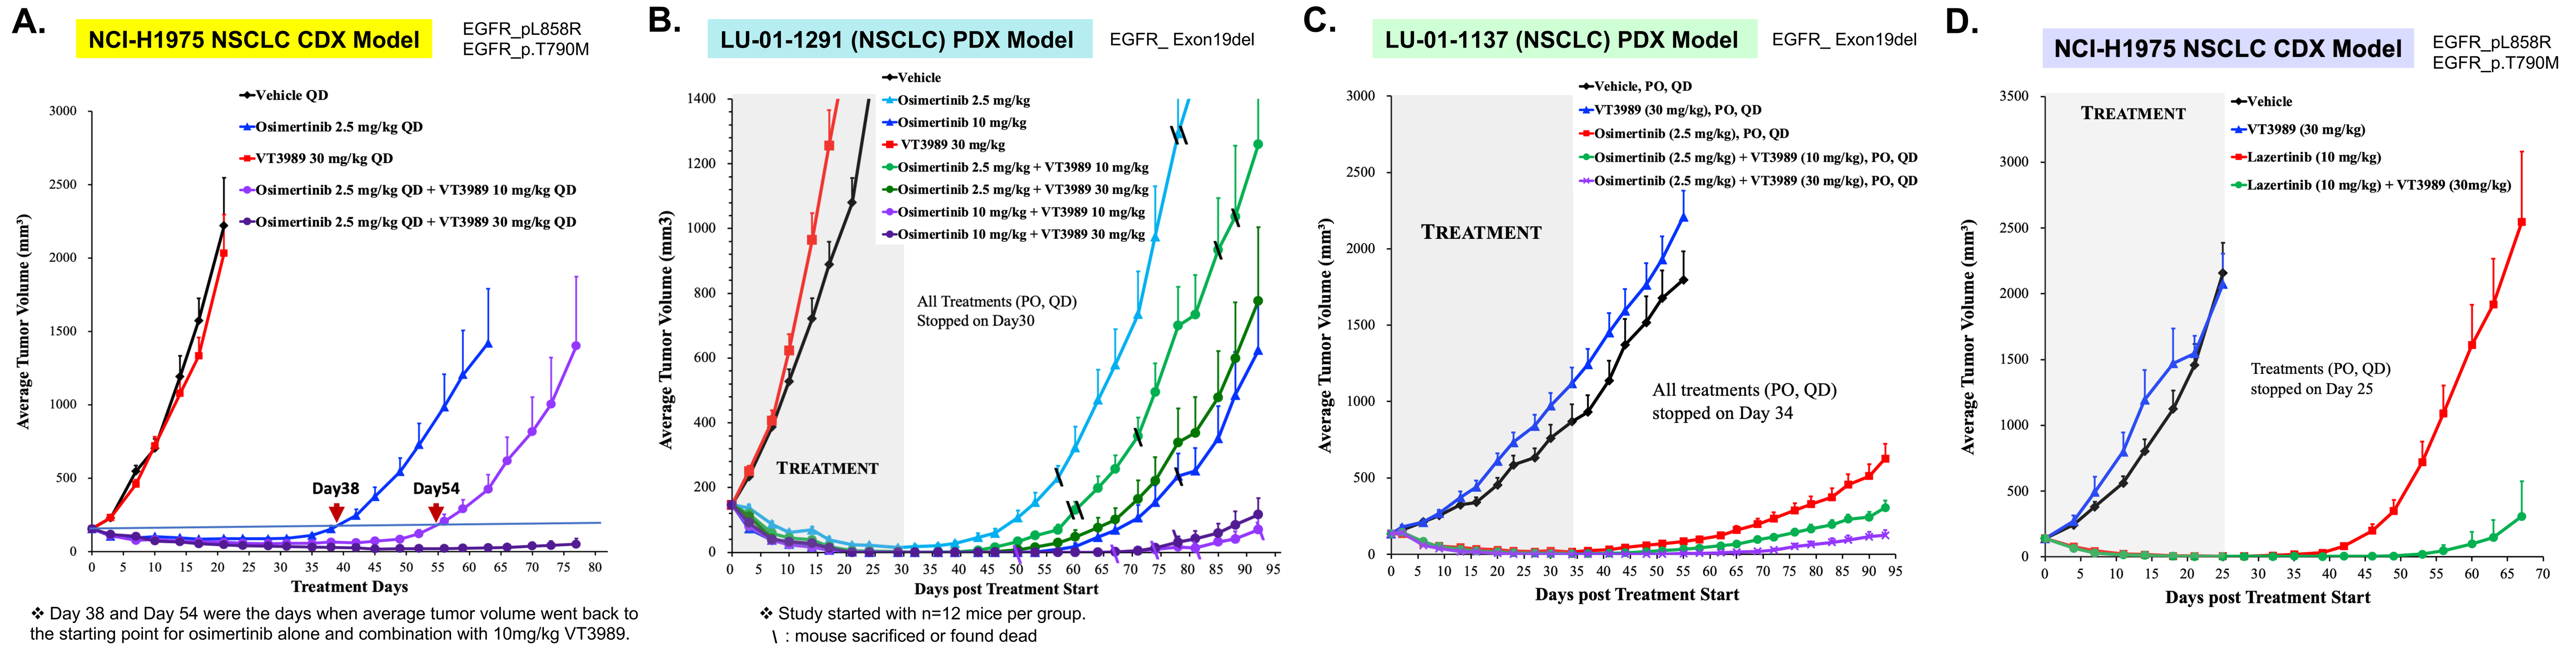

A.
NCI-H1975 NSCLC CDX Model
EGFR_pL858R
EGFR_p.T790M
Vehicle QD
Osimertinib 2.5 mg/kg QD
VT3989 30 mg/kg QD
Osimertinib 2.5 mg/kg QD + VT3989 10 mg/kg QD
Osimertinib 2.5 mg/kg QD + VT3989 30 mg/kg QD
Average Tumor Volume (mm³)
Day38
Day54
Treatment Days
❖ Day 38 and Day 54 were the days when average tumor volume went back to the starting point for osimertinib alone and combination with 10mg/kg VT3989.
B.
LU-01-1291 (NSCLC) PDX Model
EGFR_ Exon19del
Vehicle
Osimertinib 2.5 mg/kg
Osimertinib 10 mg/kg
VT3989 30 mg/kg
Osimertinib 2.5 mg/kg + VT3989 10 mg/kg
Osimertinib 2.5 mg/kg + VT3989 30 mg/kg
Osimertinib 10 mg/kg + VT3989 10 mg/kg
Osimertinib 10 mg/kg + VT3989 30 mg/kg
Average Tumor Volume (mm3)
All Treatments (PO, QD) Stopped on Day30
TREATMENT
Days post Treatment Start
❖ Study started with n=12 mice per group.
\ : mouse sacrificed or found dead
C.
LU-01-1137 (NSCLC) PDX Model
EGFR_ Exon19del
Vehicle, PO, QD
VT3989 (30 mg/kg), PO, QD
Osimertinib (2.5 mg/kg), PO, QD
Osimertinib (2.5 mg/kg) + VT3989 (10 mg/kg), PO, QD
Osimertinib (2.5 mg/kg) + VT3989 (30 mg/kg), PO, QD
TREATMENT
Average Tumor Volume (mm³)
All treatments (PO, QD) stopped on Day 34
Days post Treatment Start
D.
NCI-H1975 NSCLC CDX Model
EGFR_pL858R
EGFR_p.T790M
TREATMENT
Vehicle
VT3989 (30 mg/kg)
Lazertinib (10 mg/kg)
Lazertinib (10 mg/kg) + VT3989 (30mg/kg)
Average Tumor Volume (mm³)
Treatments (PO, QD) stopped on Day 25
Days post Treatment Start

Figure 1. VT3989 and EGFR small molecule inhibitor combination studies *in vivo*
EGFR inhibitor, osimertinib (A, B, and C) or lazertinib (D) was combined with VT3989 in EGFR mutant NSCLC CDX models (A and D) and PDX models (B and C). Treatments (>75 days) were continuous throughout the entire study in the study shown in (A). Treatments in the studies shown in (B, C, and D) were stopped at the indicated time points. Tumor volumes were measured throughout the studies to monitor tumor regrowth.VT3989 shows strong synergy in combination with EGFR inhibitors and significantly delay the tumor regrowth in combination.

Combination with KRAS-G12C inhibitor

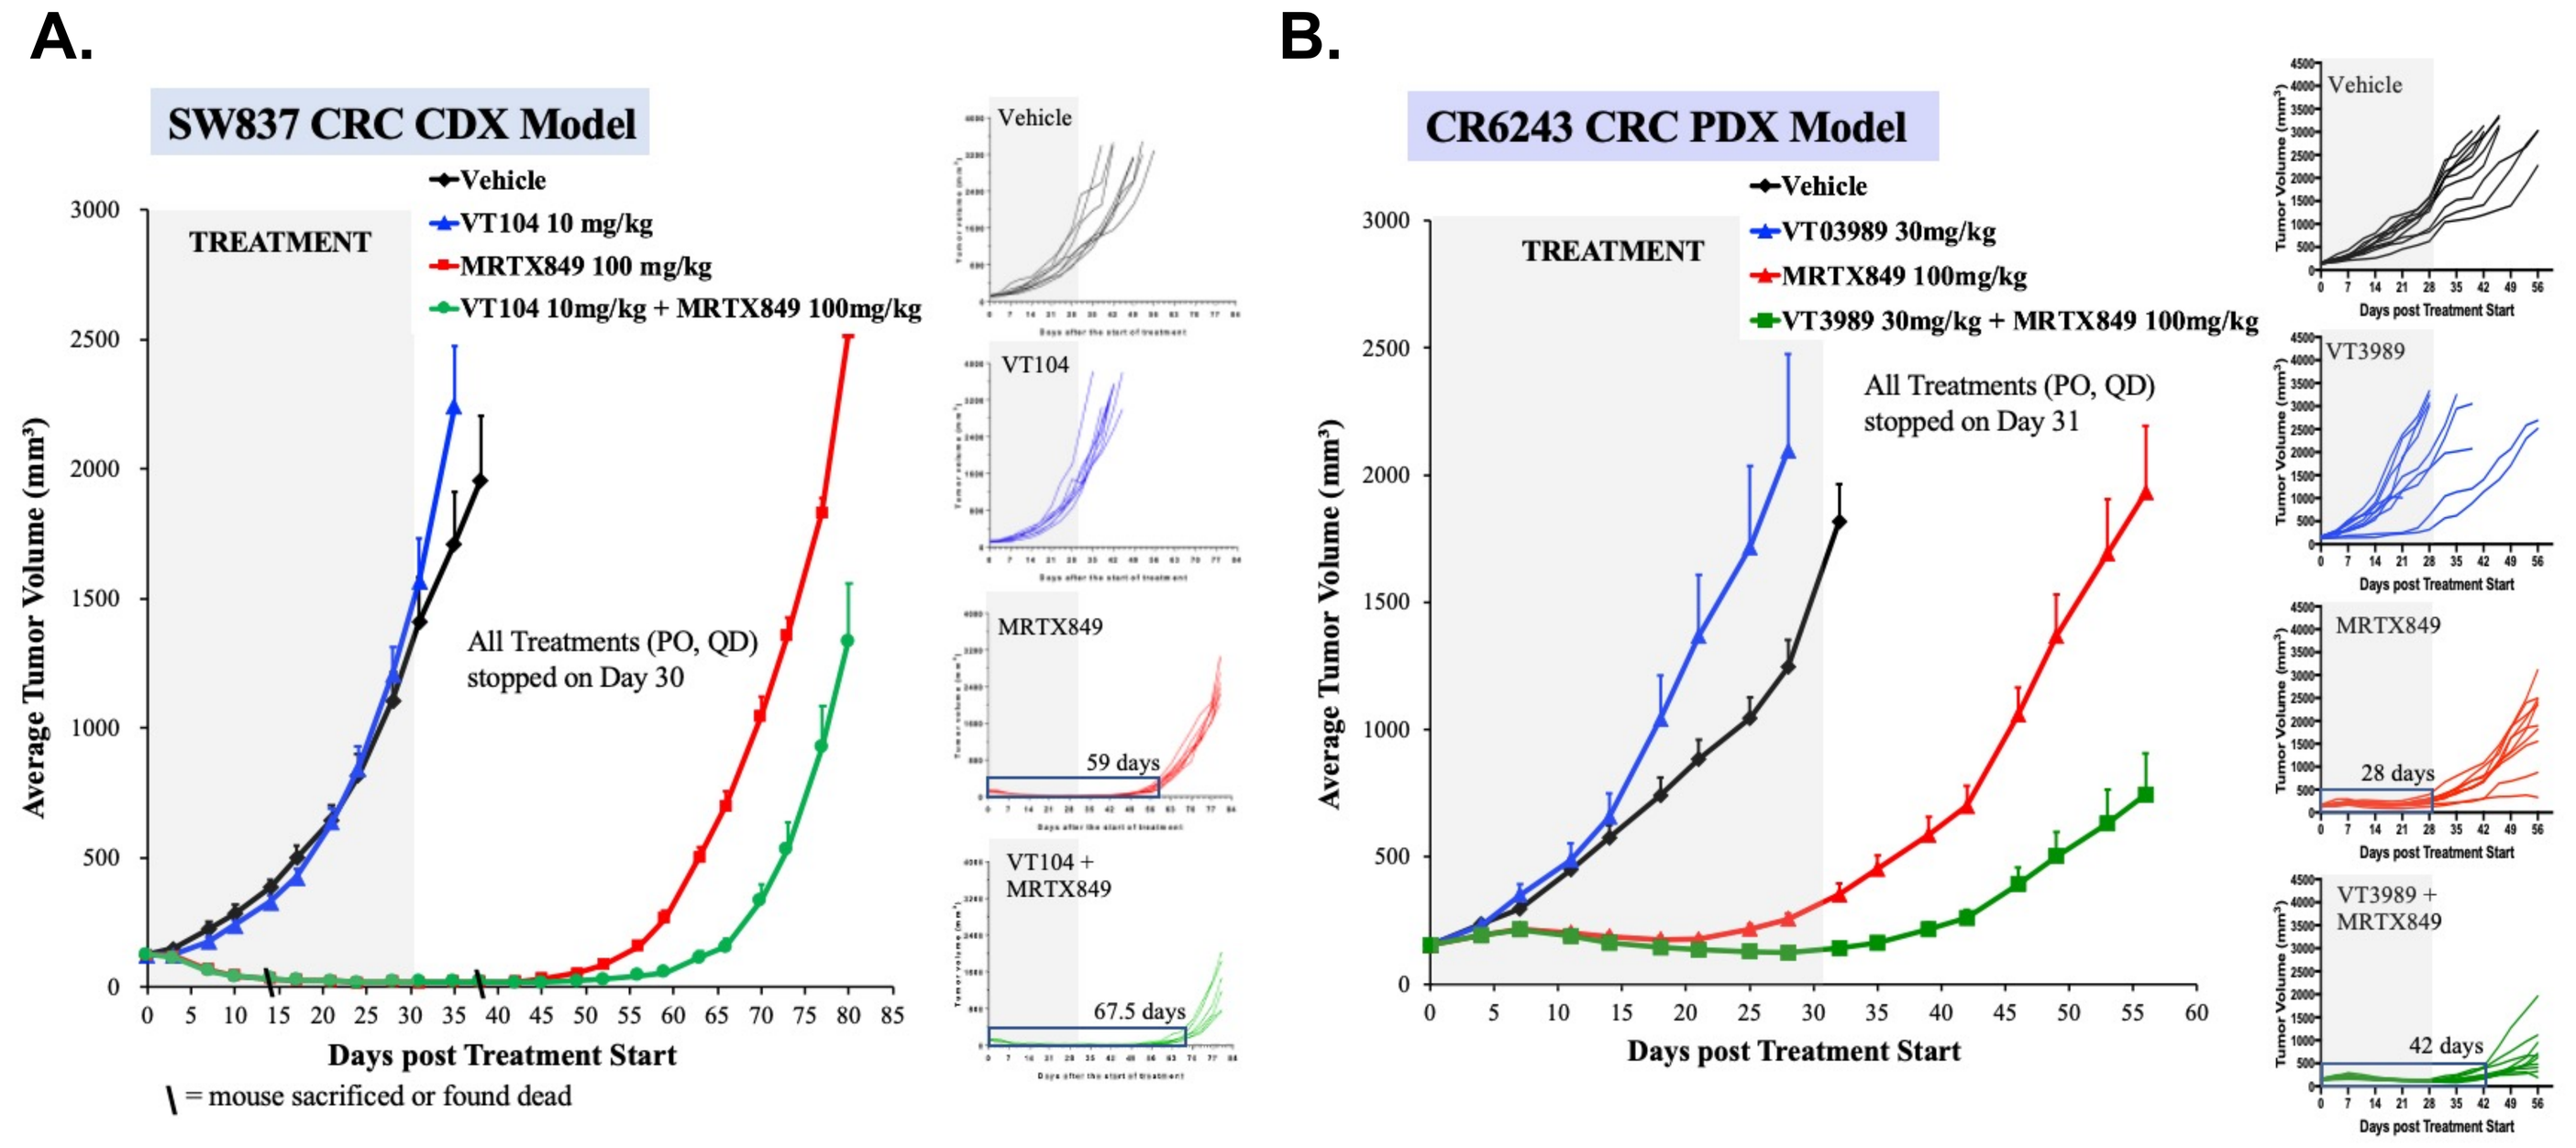

A.
SW837 CRC CDX Model
TREATMENT
Vehicle
VT104 10 mg/kg
MRTX849 100 mg/kg
VT104 10mg/kg + MRTX849 100mg/kg
Average Tumor Volume (mm³)
All Treatments (PO, QD) stopped on Day 30
Days post Treatment Start
\ = mouse sacrificed or found dead
Vehicle
VT104
MRTX849
59 days
VT104 + MRTX849
67.5 days
B.
CR6243 CRC PDX Model
TREATMENT
Vehicle
VT03989 30mg/kg
MRTX849 100mg/kg
VT3989 30mg/kg + MRTX849 100mg/kg
All Treatments (PO, QD) stopped on Day 31
Average Tumor Volume (mm³)
Days post Treatment Start
Vehicle
VT3989
MRTX849
28 days
VT3989 + MRTX849
42 days

Figure 3. TEAD and KRAS-G12C small molecule inhibitor combination studies
KRAS-G12C inhibitor, MRTX869 (adagrasib), was combined with TEAD inhibitors, VT104 (A) or VT3989 (B) in KRAS-G12C CRC models. Treatments in the studies were stopped at the indicated time points. Tumor volumes were measured throughout the studies to monitor tumor regrowth.

REFERENCE:

1. Lin et al., 2015, Nat Genet 47(3): 250-6.
2. Kurppa et al., 2020, Cancer Cell 37: 104–122.
3. Tang *et al*, 2021, *Mol Cancer Ther*. 20(6):986-998
4. Haderk et al.2021_BioRxvi_2021.10.23.465573v1
5. Tsuji et al., 2020, Nat Commun. 11(1):74.
6. Nilsson et al., 2020_Sci. Transl. Med. 12, eaaz4589

2023 AACR-NCI-EORTC Molecular Targets Conference
October 11-15, Boston, Massachusetts

Combination with BRAF / MEK inhibitors

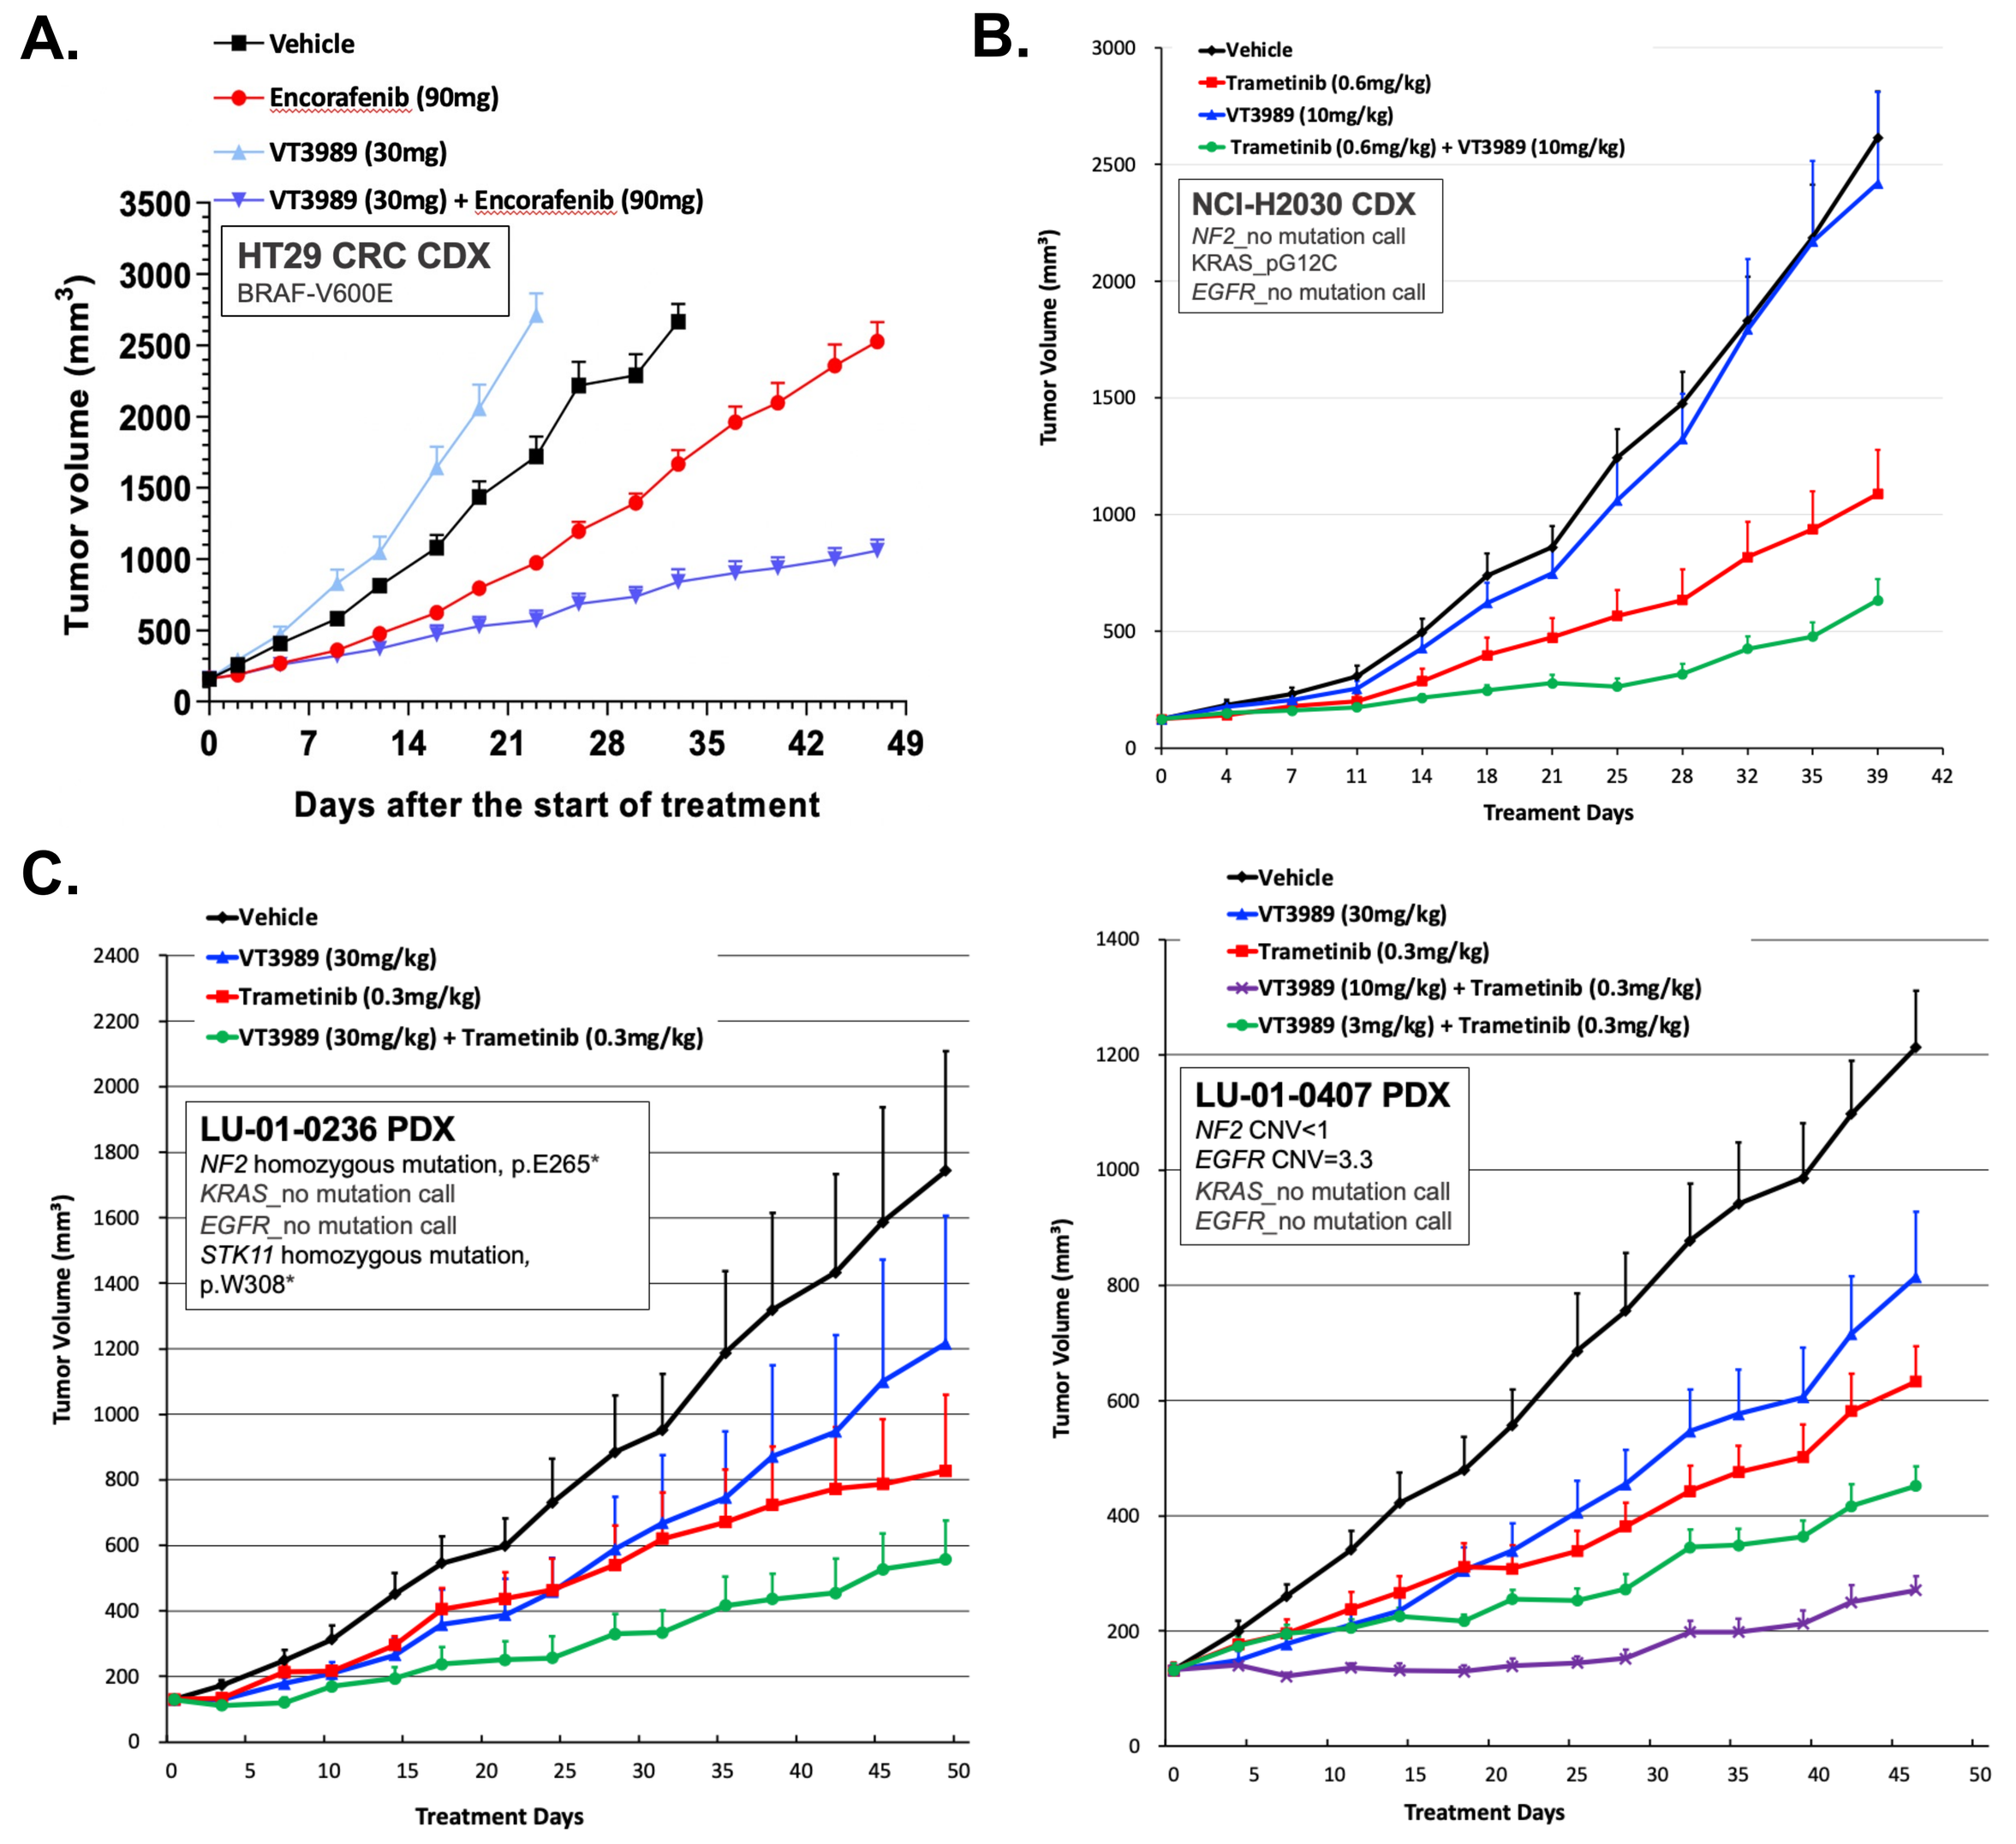

A.
Vehicle
Encorafenib (90mg)
VT3989 (30mg)
VT3989 (30mg) + Encorafenib (90mg)
HT29 CRC CDX
BRAF-V600E
Tumor volume (mm³)
Days after the start of treatment
B.
Vehicle
Trametinib (0.6mg/kg)
VT3989 (10mg/kg)
Trametinib (0.6mg/kg) + VT3989 (10mg/kg)
NCI-H2030 CDX
NF2_no mutation call
KRAS_pG12C
EGFR_no mutation call
Tumor Volume (mm³)
Treament Days
C.
Vehicle
VT3989 (30mg/kg)
Trametinib (0.3mg/kg)
VT3989 (30mg/kg) + Trametinib (0.3mg/kg)
LU-01-0236 PDX
NF2 homozygous mutation, p.E265*
KRAS_no mutation call
EGFR_no mutation call
STK11 homozygous mutation, p.W308*
Tumor Volume (mm³)
Treatment Days
Vehicle
VT3989 (30mg/kg)
Trametinib (0.3mg/kg)
VT3989 (10mg/kg) + Trametinib (0.3mg/kg)
VT3989 (3mg/kg) + Trametinib (0.3mg/kg)
LU-01-0407 PDX
NF2 CNV<1
EGFR CNV=3.3
KRAS_no mutation call
EGFR_no mutation call
Tumor Volume (mm³)
Treatment Days

Figure 4. VT3989 and BRAF / MEK small molecule inhibitors combination studies
(A) VT3989 shows strong synergy with BRAF inhibitor encorafenib in HT29 CRC CDX model. (B and C) VT3989 shows combination effect with trametinib in NSCLC CDX (B) and NSCLC PDX models (C). All treatments were PO, QD throughout the studies.

Combination with EGFR-MET bispecific antibody +/- EGFR small molecule inhibitor

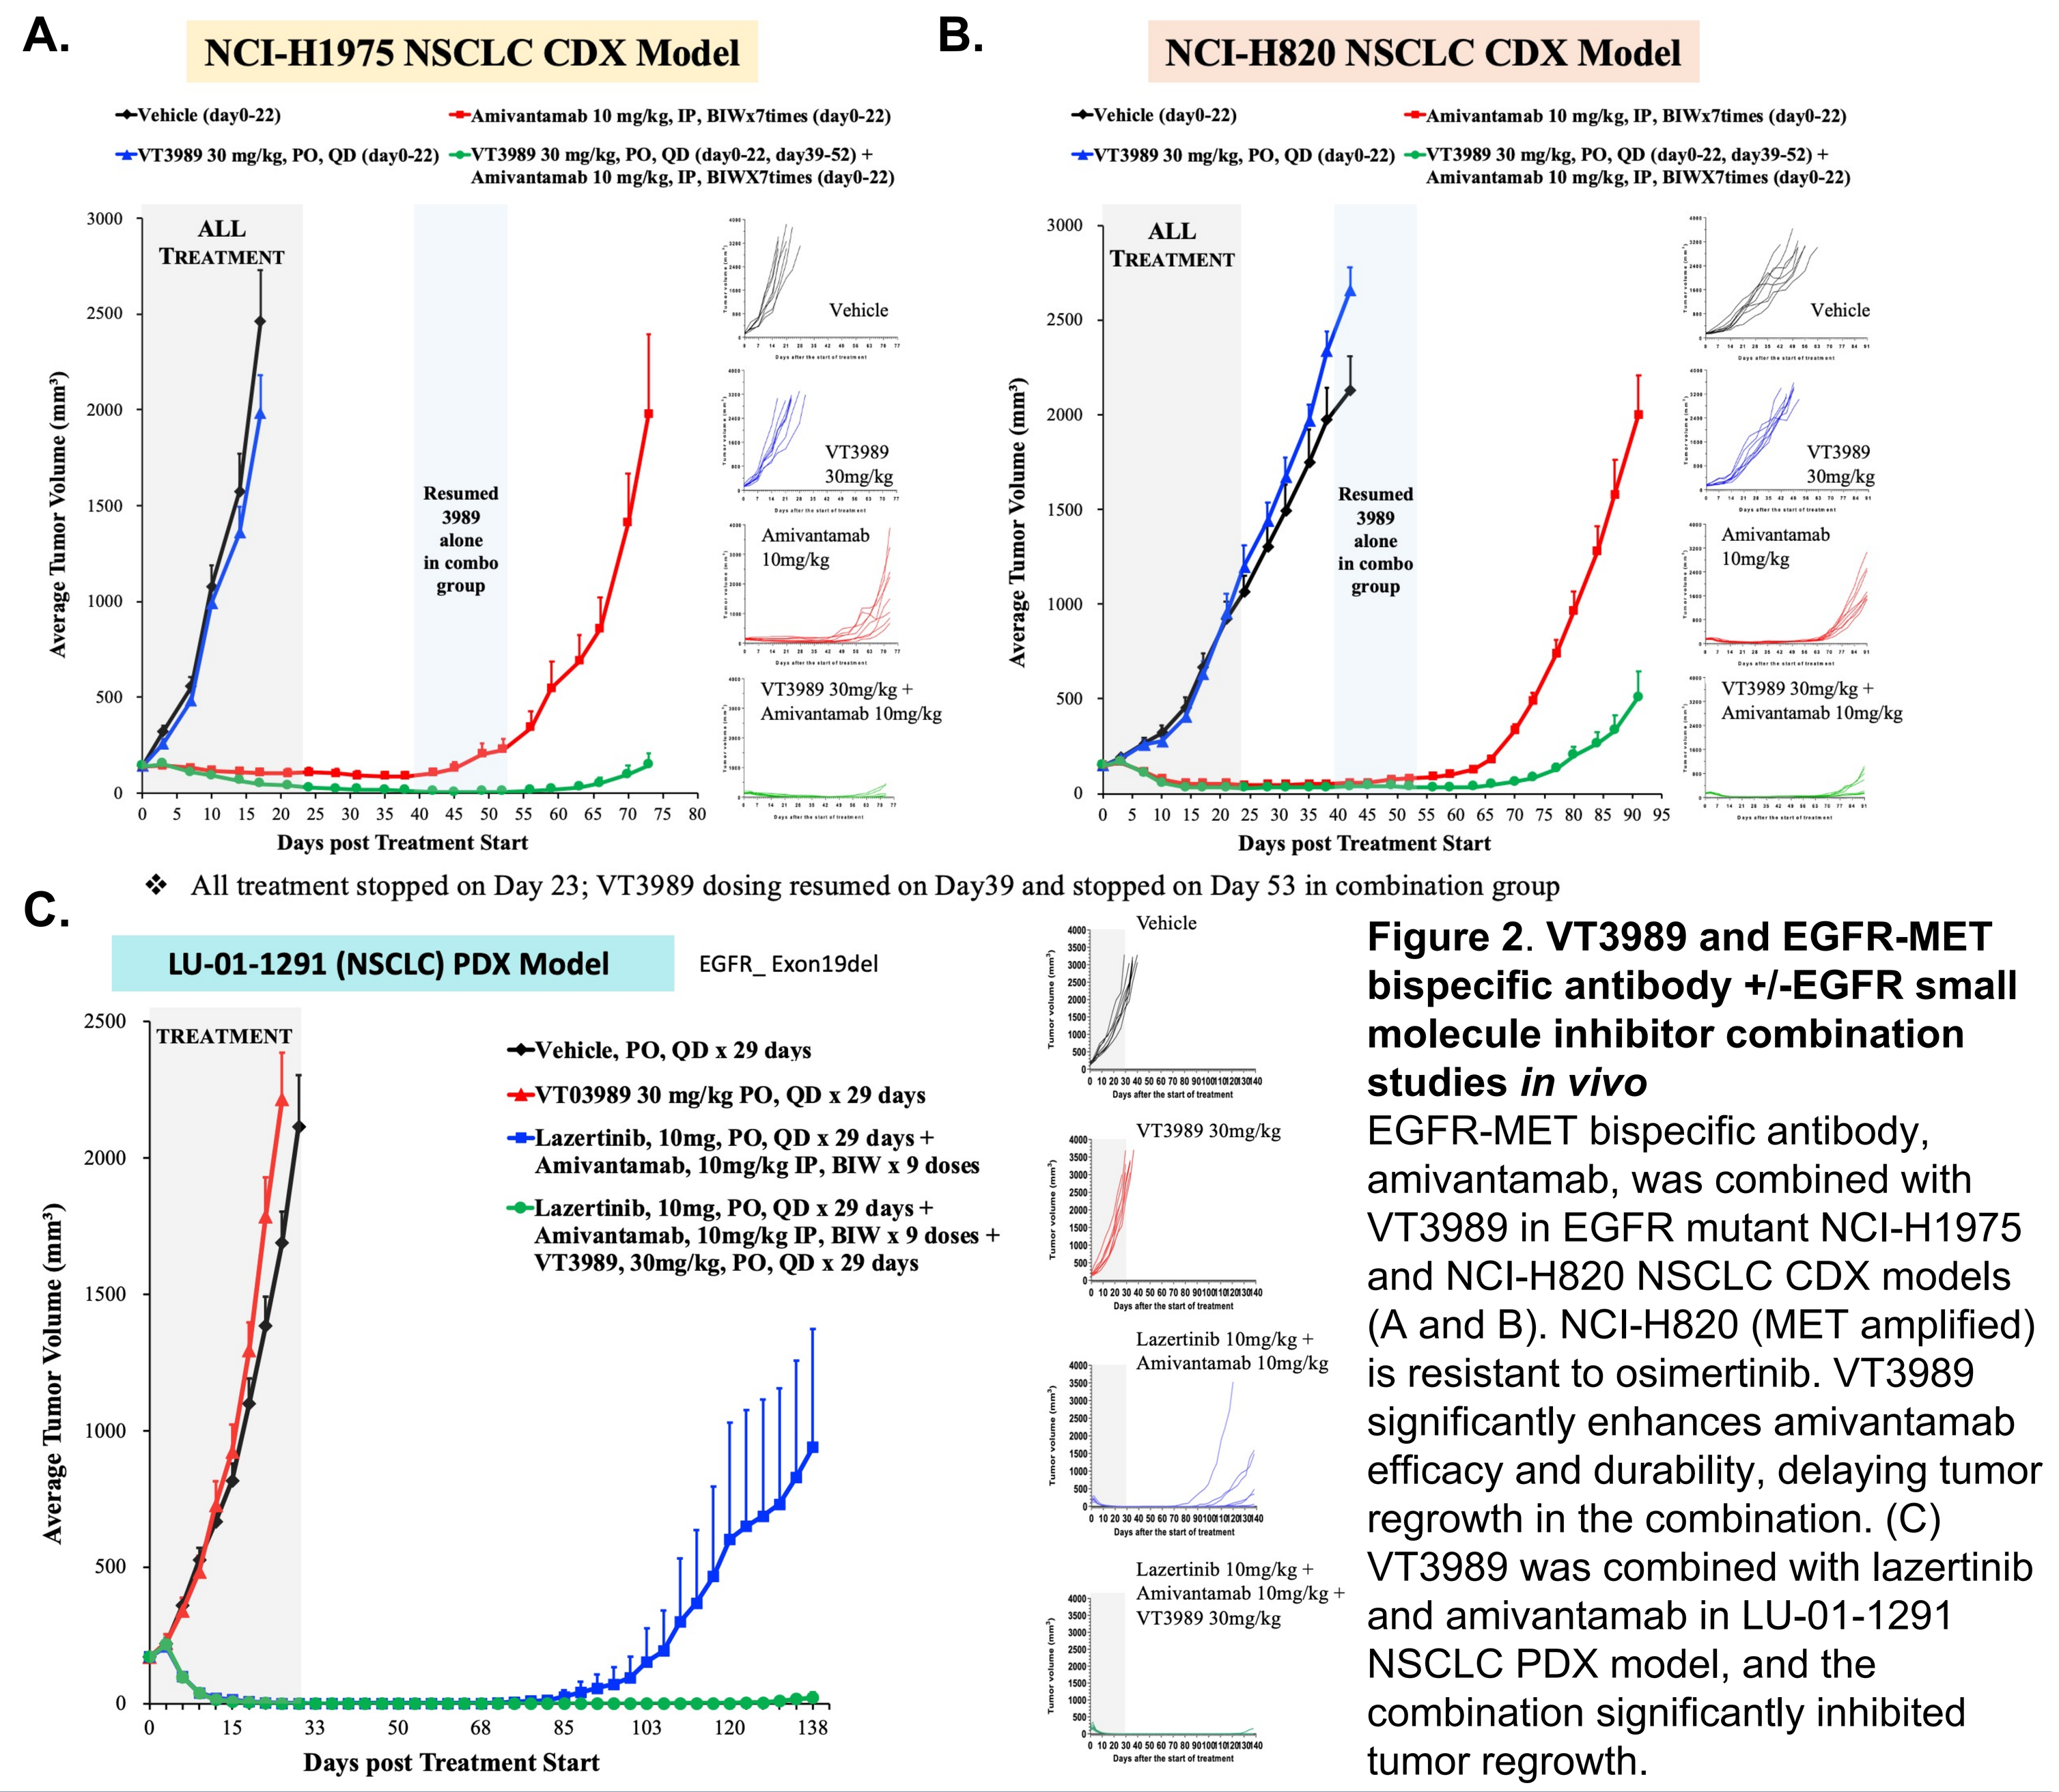

A.
NCI-H1975 NSCLC CDX Model
Vehicle (day0-22)
Amivantamab 10 mg/kg, IP, BIWx7times (day0-22)
VT3989 30 mg/kg, PO, QD (day0-22)
VT3989 30 mg/kg, PO, QD (day0-22, day39-52) + Amivantamab 10 mg/kg, IP, BIWX7times (day0-22)
ALL TREATMENT
Resumed 3989 alone in combo group
Average Tumor Volume (mm³)
Days post Treatment Start
Vehicle
VT3989 30mg/kg
Amivantamab 10mg/kg
VT3989 30mg/kg + Amivantamab 10mg/kg
B.
NCI-H820 NSCLC CDX Model
❖ All treatment stopped on Day 23; VT3989 dosing resumed on Day39 and stopped on Day 53 in combination group
C.
LU-01-1291 (NSCLC) PDX Model
EGFR_ Exon19del
TREATMENT
Vehicle, PO, QD x 29 days
VT03989 30 mg/kg PO, QD x 29 days
Lazertinib, 10mg, PO, QD x 29 days + Amivantamab, 10mg/kg IP, BIW x 9 doses
Lazertinib, 10mg, PO, QD x 29 days + Amivantamab, 10mg/kg IP, BIW x 9 doses + VT3989, 30mg/kg, PO, QD x 29 days
Days post Treatment Start
Lazertinib 10mg/kg + Amivantamab 10mg/kg
Lazertinib 10mg/kg + Amivantamab 10mg/kg + VT3989 30mg/kg

Figure 2. VT3989 and EGFR-MET bispecific antibody +/-EGFR small molecule inhibitor combination studies *in vivo*
EGFR-MET bispecific antibody, amivantamab, was combined with VT3989 in EGFR mutant NCI-H1975 and NCI-H820 NSCLC CDX models (A and B). NCI-H820 (MET amplified) is resistant to osimertinib. VT3989 significantly enhances amivantamab efficacy and durability, delaying tumor regrowth in the combination. (C) VT3989 was combined with lazertinib and amivantamab in LU-01-1291 NSCLC PDX model, and the combination significantly inhibited tumor regrowth.

Combination with MET small molecule inhibitor

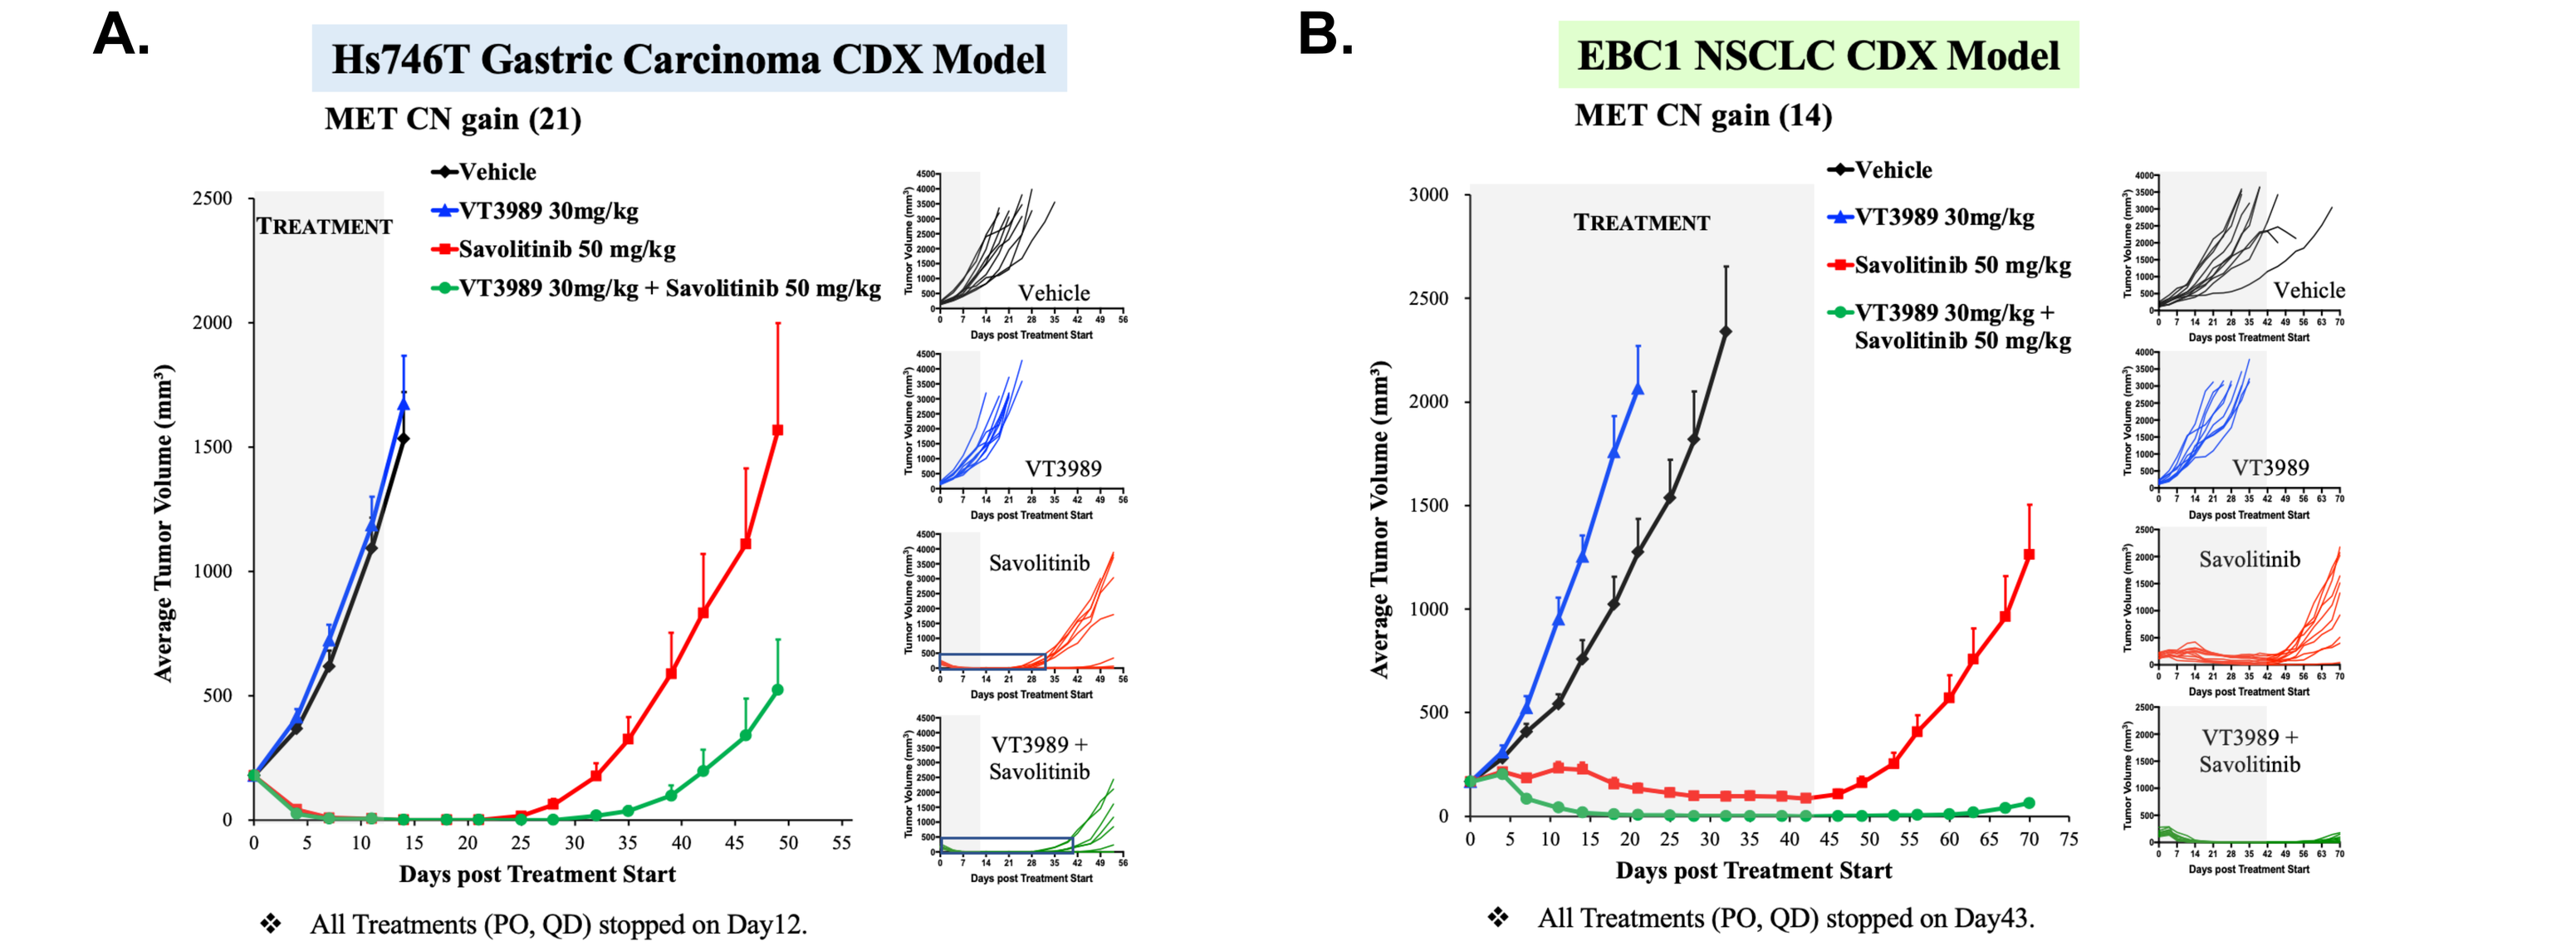

A.
Hs746T Gastric Carcinoma CDX Model
MET CN gain (21)
TREATMENT
Vehicle
VT3989 30mg/kg
Savolitinib 50 mg/kg
VT3989 30mg/kg + Savolitinib 50 mg/kg
Average Tumor Volume (mm³)
Days post Treatment Start
❖ All Treatments (PO, QD) stopped on Day12.
B.
EBC1 NSCLC CDX Model
MET CN gain (14)
❖ All Treatments (PO, QD) stopped on Day43.

Figure 5. VT3989 and MET inhibitor combination studies *in vivo*
MET inhibitor, savolitinib, was combined with VT3989 in MET amplified CDX models. VT3989 significantly enhances savolitinib efficacy and durability, delaying tumor regrowth in the combination.

Combination with mTOR small molecule inhibitor

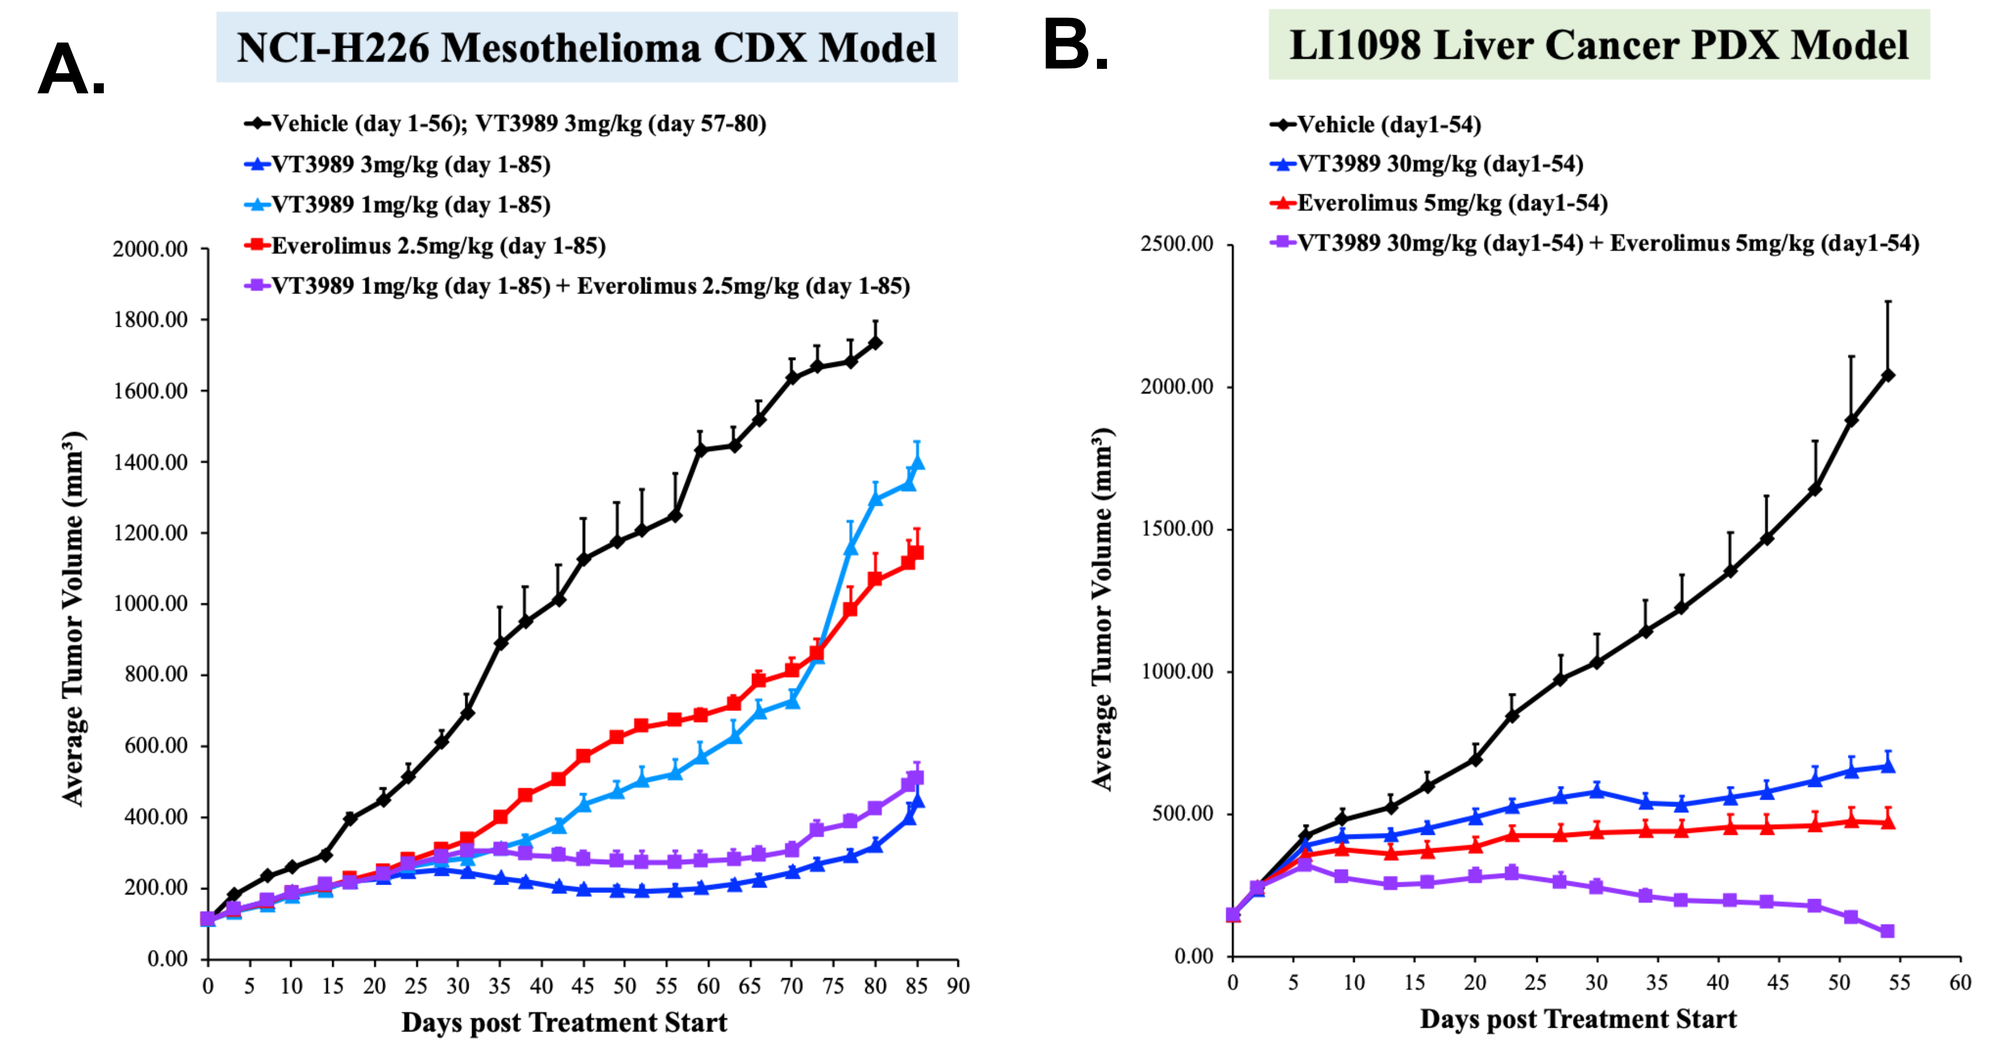

A.
NCI-H226 Mesothelioma CDX Model
Vehicle (day 1-56); VT3989 3mg/kg (day 57-80)
VT3989 3mg/kg (day 1-85)
VT3989 1mg/kg (day 1-85)
Everolimus 2.5mg/kg (day 1-85)
VT3989 1mg/kg (day 1-85) + Everolimus 2.5mg/kg (day 1-85)
Average Tumor Volume (mm³)
Days post Treatment Start
B.
LI1098 Liver Cancer PDX Model
Vehicle (day1-54)
VT3989 30mg/kg (day1-54)
Everolimus 5mg/kg (day1-54)
VT3989 30mg/kg (day1-54) + Everolimus 5mg/kg (day1-54)

Figure 6. VT3989 and mTOR inhibitor combination studies in vivo
Everolimus mTOR inhibitor was combined with VT3989 in mesothelioma CDX model (A) and liver cancer PDX model (B). VT3989 shows strong synergy with everolimus. Treatments were administered continuously throughout the entire studies.