First-in-human phase 1 trial of VT3989, a first-in-class YAP/TEAD inhibitor in patients with advanced mesothelioma

Timothy A. Yap[1], Jayesh Desai[2], Ibiayi Dagogo-Jack[3], Michael Millward[4], Hedy L. Kindler[5], Anthony W. Tolcher[6], Sophia Frentzas[7], Amy L. Body[7], Archie Thurston[8], Len Post[9], F. Andrew Dorr[9], David J. Kwiatkowski[10]

[1]The University of Texas MD Anderson Cancer Center, Houston, TX, USA, [2]Peter MacCallum Cancer Centre, Melbourne, AU, [3]Massachusetts General Hospital Cancer Center, Boston, MA, USA, [4]Linear Cancer Trials, Queen Elizabeth II Medical Centre, Perth, AU, [5]University of Chicago, Chicago, IL, USA, [6]NEXT Oncology, San Antonio, TX, USA, [7]Monash Medical Centre, Clayton, AU, [8]Toxicology Solutions, Marana, AZ, USA, [9]Vivace Therapeutics, San Mateo, CA, USA, [10]Brigham and Women's Hospital & Dana Farber Cancer Institute, Boston, MA,USA

Background

VT3989 and the Hippo-Yap/TAZ-TEAD signaling pathway

Hippo signaling regulates transcription factors YAP and TAZ in response to diverse upstream signals

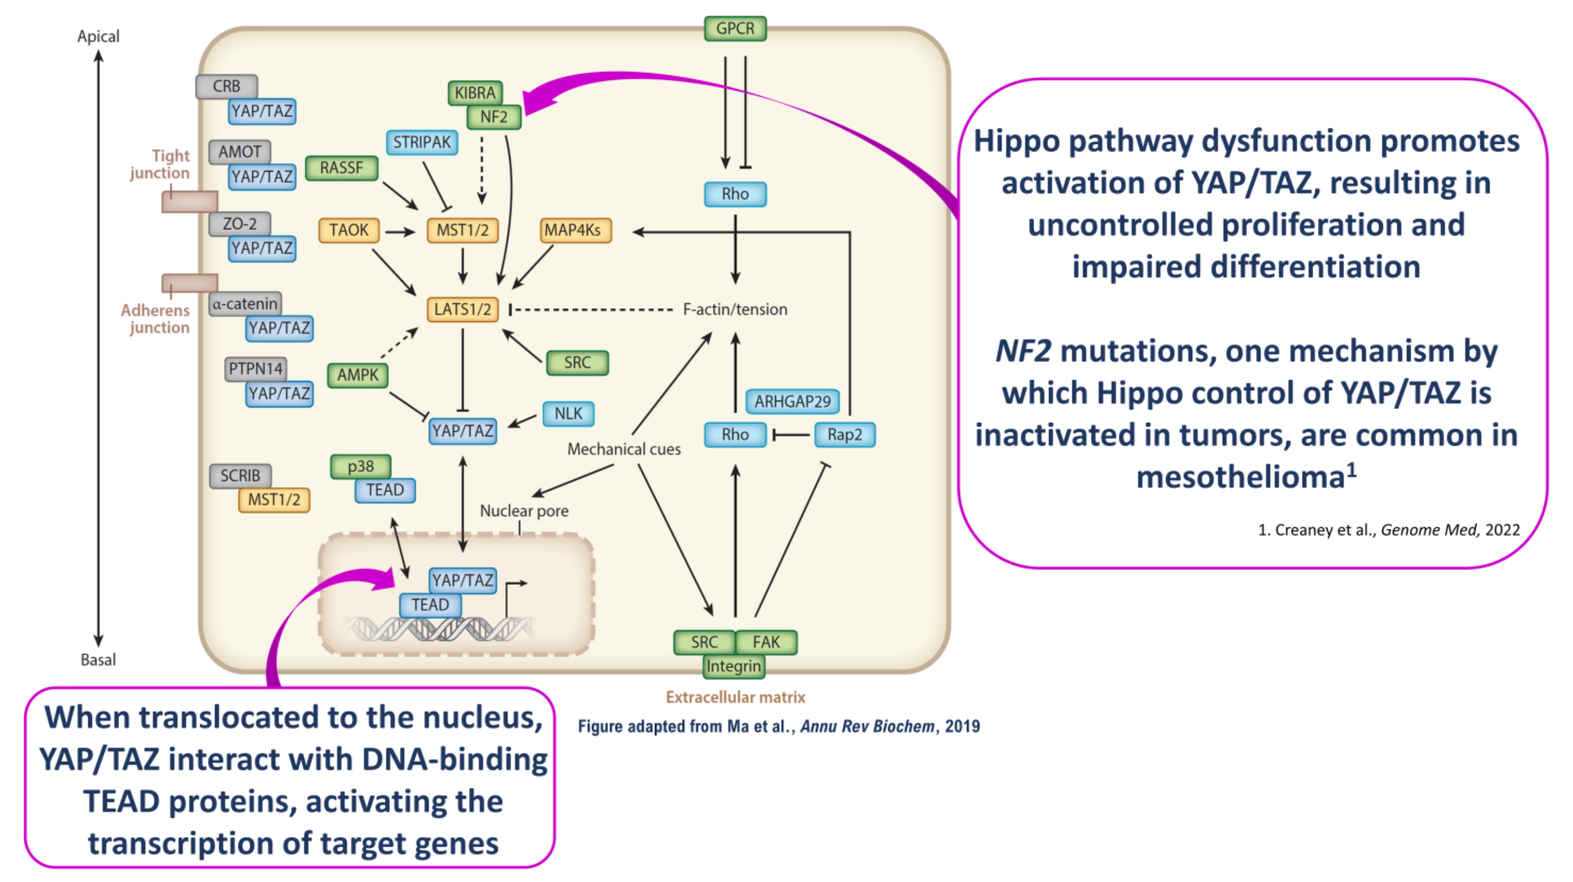
Hippo pathway dysfunction promotes activation of YAP/TAZ, resulting in uncontrolled proliferation and impaired differentiation
NF2 mutations, one mechanism by which Hippo control of YAP/TAZ is inactivated in tumors, are common in mesothelioma[1]
1. Creaney et al., Genome Med, 2022
When translocated to the nucleus, YAP/TAZ interact with DNA-binding TEAD proteins, activating the transcription of target genes
Figure adapted from Ma et al., Annu Rev Biochem, 2019

Palmitoylation of a conserved cysteine is required for TEAD interaction with YAP

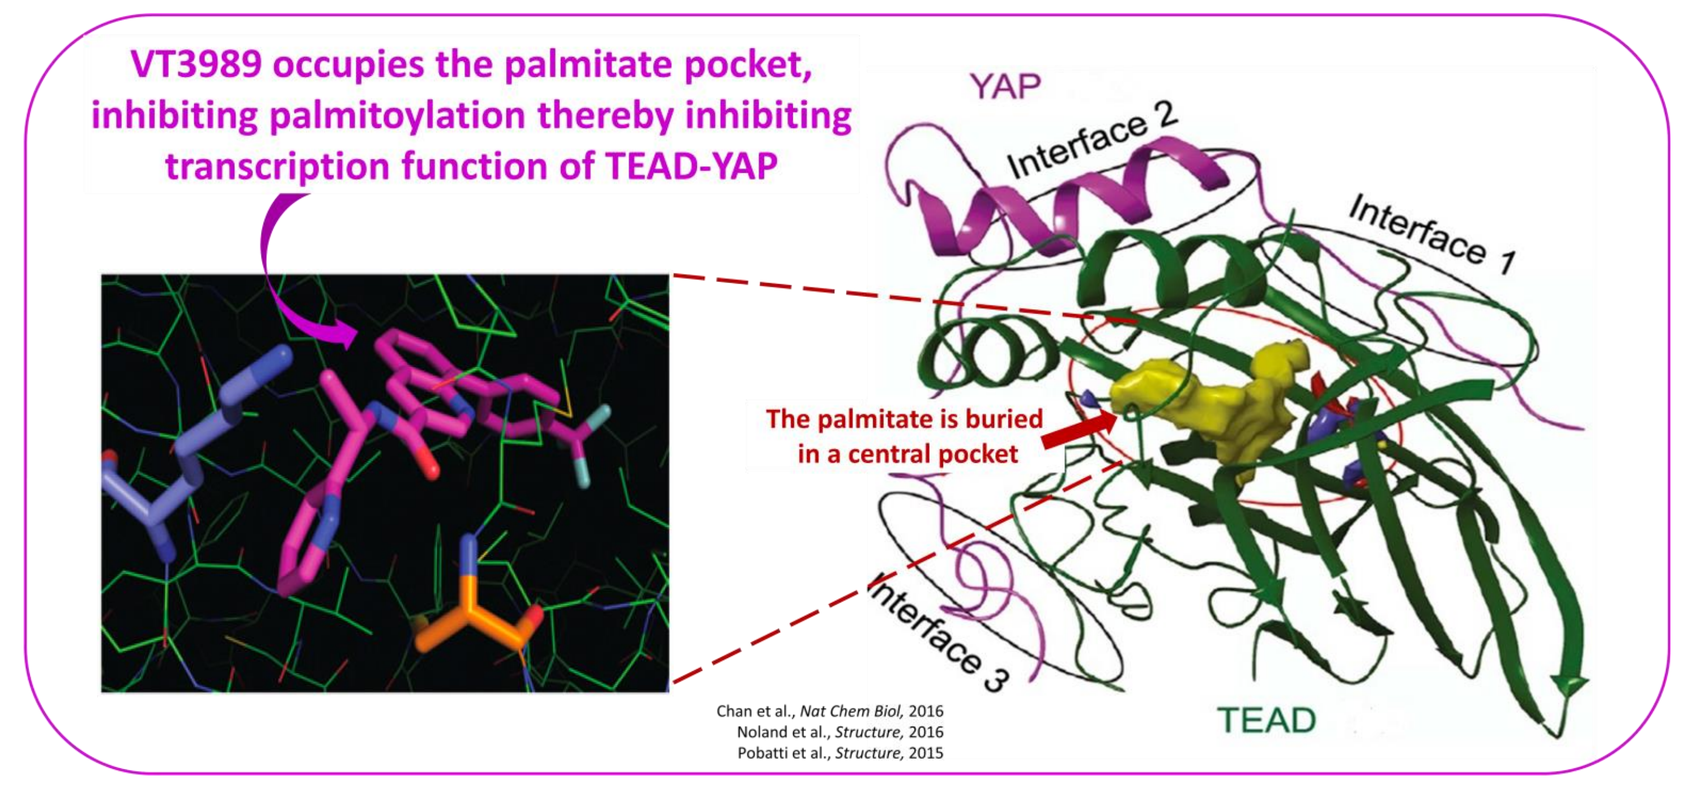
VT3989 occupies the palmitate pocket, inhibiting palmitoylation thereby inhibiting transcription function of TEAD-YAP
YAP
Interface 2
Interface 1
Interface 3
The palmitate is buried in a central pocket
TEAD
Chan et al., Nat Chem Biol, 2016
Noland et al., Structure, 2016
Pobatti et al., Structure, 2015

First-in class, first-in human Phase 1 Study of VT3989

Study Design
- Advanced refractory solid tumors, enriched for malignant pleural mesothelioma and other tumors with *NF2* mutations
- 3+3 dose escalation with dose expansion
- 3 or 4-week treatment cycles
- Response assessment every 8 to 9 weeks
- Key inclusion criteria:
 - ECOG PS 0-2
 - hemoglobin ≥ 8 g/dL, ANC ≥ 1.5 K/uL, platelets ≥ 100 K/uL, ALT/AST ≤ 2.5 x ULN, bilirubin ≤ 1.5 mg/dL, creatinine ≤ ULN, estimated GFR ≥ 60 mL/min if creatinine 1-1.5 x ULN, serum albumin > 2.5 g/dL, UACR* ≤ 100 mg/gm

Study Endpoints

Primary:
- Safety and tolerability
- Maximum Tolerated Dose (MTD) and Recommended Phase 2 Dose (RP2D)

Secondary:
- Preliminary antitumor activity in *NF2* mutation tumors
- Pharmacokinetics
- Time to response and duration of response

Exploratory:
- Hippo-Yap signaling in sequential tumor biopsies
- ctDNA changes

Dose cohorts and escalation schema, n = all patients [] = mesothelioma patients

Continuous

Cohort	Dose	n
1	25 mg QD	n = 5 [1] / 0 DLT
2	50 QD	n = 6 [4]/ 0 DLT
3	100 mg QD	n = 6 [3]/ 0 DLT
4	150 mg QD	n = 6 [4] / 0 DLT
5	200 mg QD	n = 4 [1]/ 0 DLT

Though no Cycle 1 DLTs occurred, proteinuria developed in later cycles leading to modified doses and schedules being evaluated

Intermittent

Cohort	Dose	n
6	200 mg 2 weeks on/1 week off	n = 6 [3]/ 0 DLT
7	200 mg 1 weeks on/2 weeks off	n = 6 [4]/ 0 DLT
8	200 mg 2 weeks on/2 weeks off	n = 6 [2]/ 0 DLT
9	100 mg 2 weeks on/2 weeks off	n = 6 [5]/ 0 DLT
10	200 mg 2 weeks on/1 week off	n = 6 [4]/ 0 DLT
11	100 mg x 15 QD, then weekly (D1, D8, D15)	n = 7 [7]/ 0 DLT
12	50 mg x 15 QD, then weekly (D1, D8, D15)	n = 6 [6]/ 0 DLT

VT3989 Pharmacokinetics

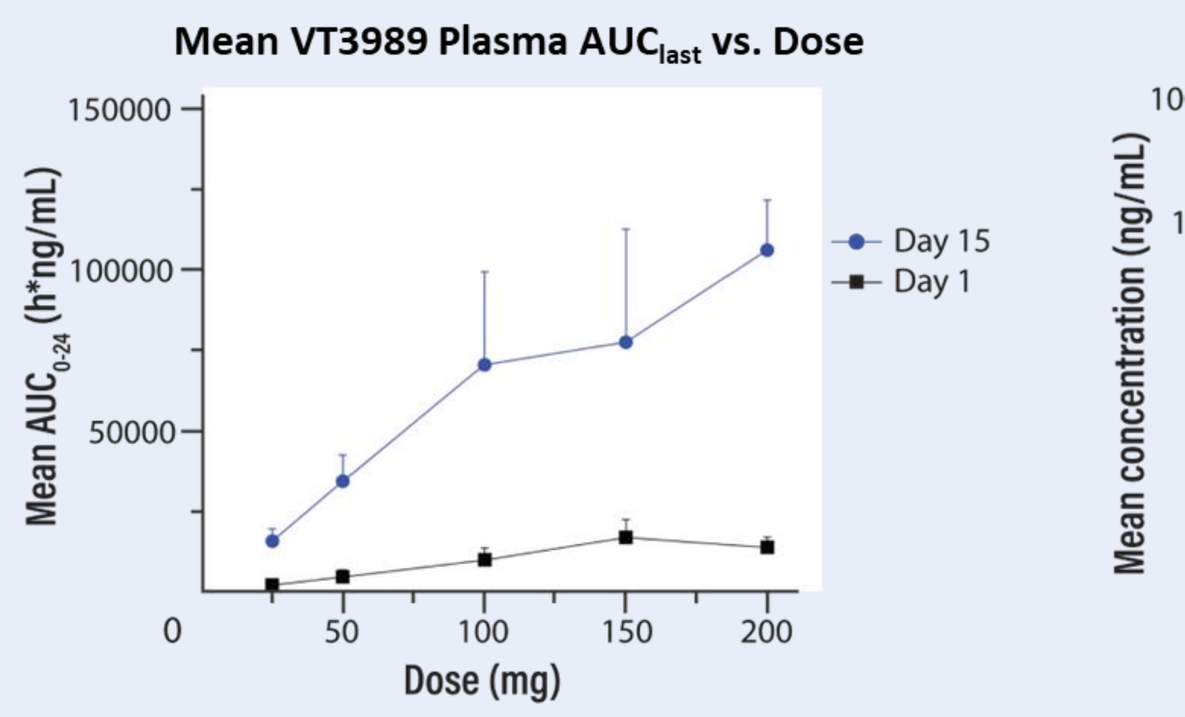
Mean VT3989 Plasma AUClast vs. Dose
Mean AUC0-24 (h*ng/mL)
Dose (mg)
Day 15
Day 1

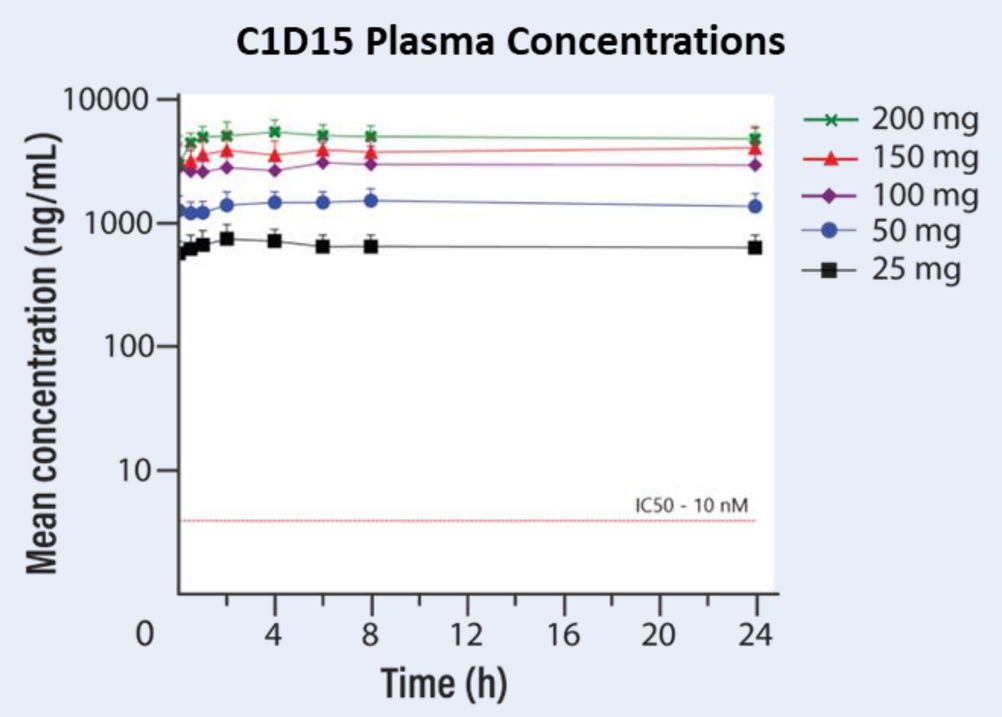
C1D15 Plasma Concentrations
Mean concentration (ng/mL)
Time (h)
200 mg
150 mg
100 mg
50 mg
25 mg
IC50 - 10 nM

- Dose-proportional exposure following oral administration
- Long half-life (approximately 12-15 days)
- Exposure on Day 15 is approximately 7x that of Day 1

First early clinical proof-of-concept that VT3989 inhibition of the Hippo-YAP-TEAD signaling pathway has antitumor efficacy

Durable activity seen in patients with advanced mesothelioma +/- *NF2* mutations

- Intermittent dosing of VT3989 at ≤100 mg results in less frequent/severe albuminuria which is reversible with dose reduction or interruption
- Dose-optimization expansion cohorts currently evaluating different doses/schedules in 2-stage designs
- Multivariate analyses underway to explore potential predictors of nephropathy

Baseline demographics and characteristics – mesothelioma patients

Baseline Demographics	N = 44 (%)
Median Age (range)	63.3 (21 - 83)
Gender	
Female (%)	19 (43)
Male (%)	25 (57)
Race	
White (%)	38 (86.4)
Black (%)	2 (4.5)
Other (%)	4 (9.1)
Ethnicity	
Hispanic (%)	4 (9.1)
ECOG Performance Status	
0 (%)	6 (13.6)
1 (%)	38 (86.4)

Baseline Characteristic	N = 44 (%)
Tumor Type	
Pleural	32 (72.7)
Peritoneal	10 (22.7)
Pericardial	1 (2.2)
Bicavitary	1 (2.2)
Molecular Profile	
NF2 Mutation	19 (43.2)
NF2 Wildtype	9 (20.5)
Unknown	16 (36.4)
Histology	
Epithelioid	35 (79.5)
Sarcomatoid	2 (4.5)
Biphasic/mixed	7 (15.9)
Prior Therapy (> 10 Patients)	
Median (Range)	3 (1 - 8)
Platinum-pemetrexed (%)	43 (98)
Immune checkpoint inhibitor (%)	39 (89)
Anti-VEGF antibody (%)	21 (48)
Gemcitabine (%)	11 (25)
Vinorelbine (%)	11 (25)

Results

Safety

Treatment-related adverse events in ≥ 5 patients (n = 44)*

Adverse Event	CTCAE Grade 1 n	2 n	3 n	4 n	Total n (%)
Albuminuria	11	14	2	0	27 (61.6)
Proteinuria	14	12	0	0	26 (59.1)
Fatigue	9	4	1	0	14 (31.8)
Peripheral edema	10	3	0	0	13 (29.5)
Nausea	8	1	0	0	9 (20.5)
Increased ALT	4	1	1	0	6 (13.6)
Increased AST	4	1	1	0	6 (13.6)
Anemia	2	3	0	0	5 (11.4)
Decreased appetite	5	0	0	0	5 (11.4)

Albuminuria

	UACR (mg/gm) Cohorts 1-8, 10 (n = 26)	Cohorts 9, 11, 12 (n = 18)
Median (min, max)	255.7 (17.0, 2715.4)	86.4 (2.94, 2000.0)
Mean (SD)	654.3 (758.3)	297.8 (517.6)

- Albuminuria/proteinuria have not been associated with significant clinical symptoms or changes in serum creatinine, creatinine clearance, or serum albumin

Time on study and antitumor activity

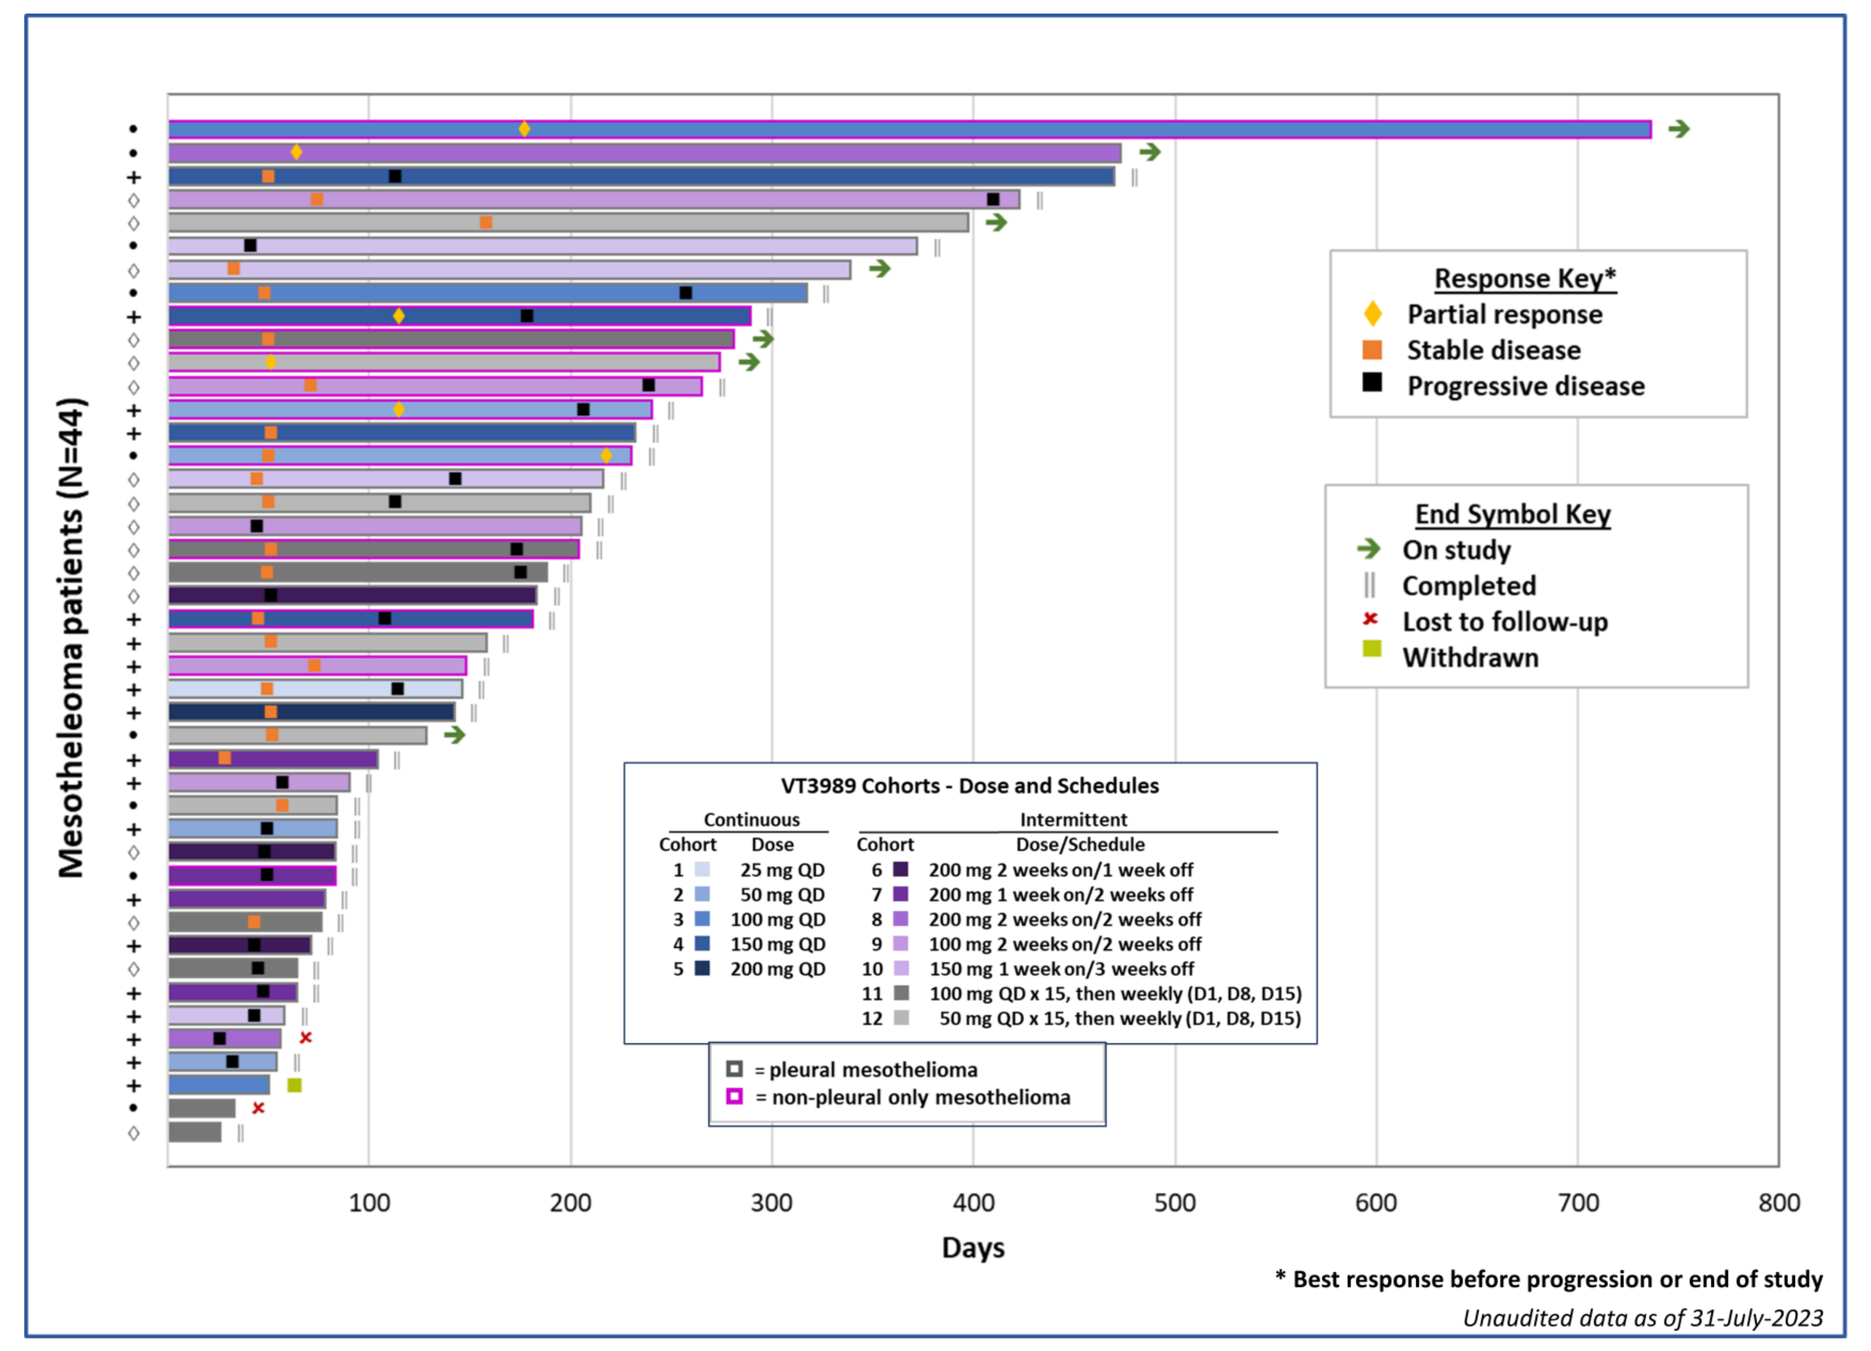
Mesotheleoma patients (N=44)
Days
Response Key*
Partial response
Stable disease
Progressive disease
End Symbol Key
On study
Completed
Lost to follow-up
Withdrawn
VT3989 Cohorts - Dose and Schedules
= pleural mesothelioma
= non-pleural only mesothelioma
* Best response before progression or end of study
Unaudited data as of 31-July-2023

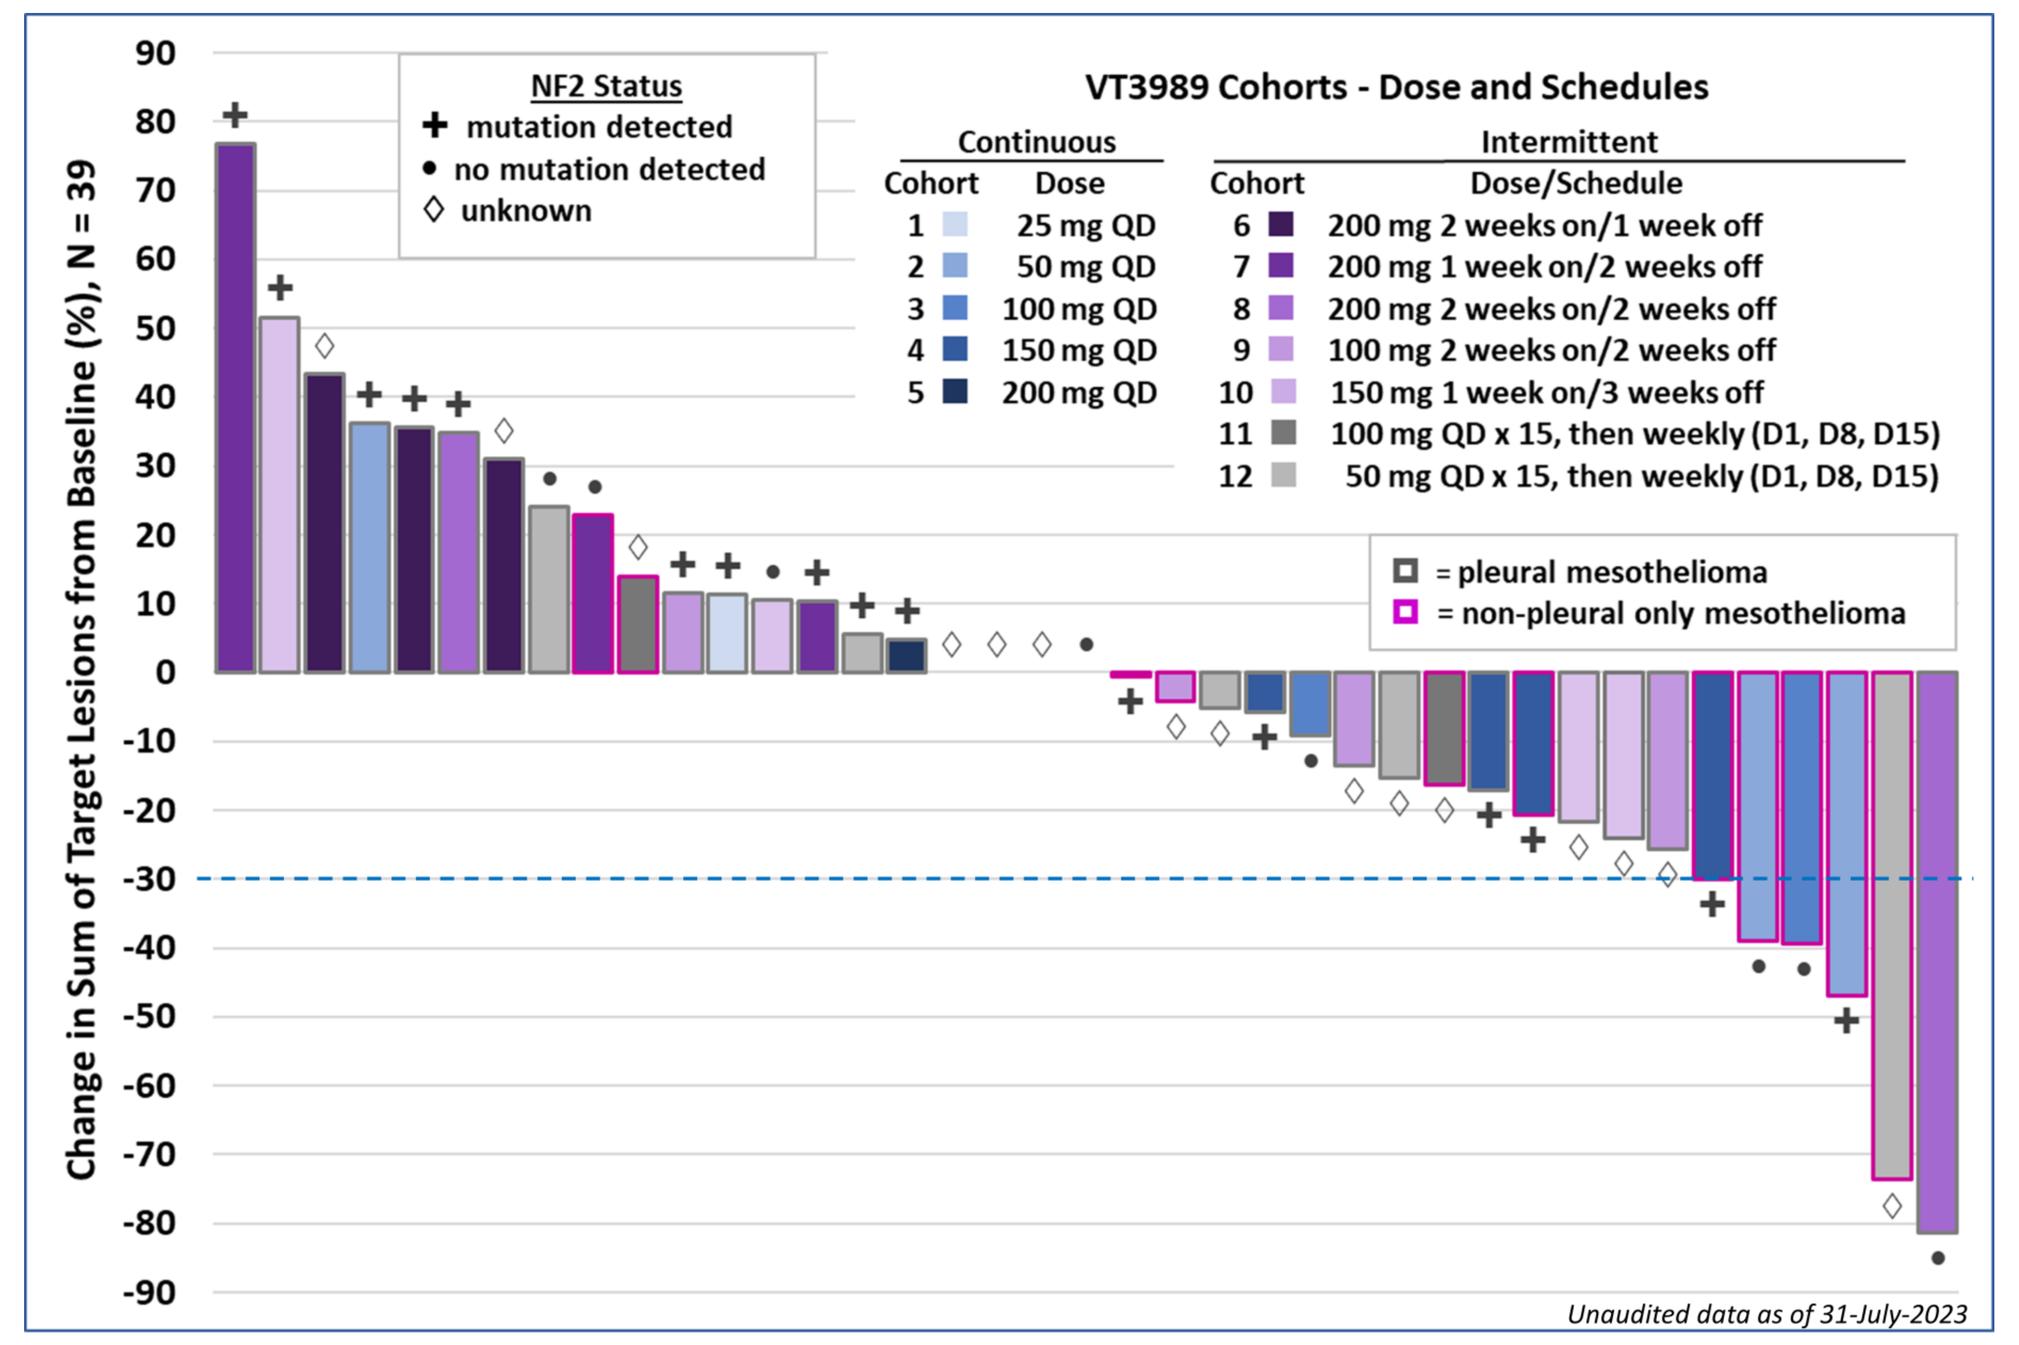
Change in Sum of Target Lesions from Baseline (%), N = 39
NF2 Status
mutation detected
no mutation detected
unknown
VT3989 Cohorts - Dose and Schedules
Continuous
Cohort Dose
1 25 mg QD
2 50 mg QD
3 100 mg QD
4 150 mg QD
5 200 mg QD
Intermittent
Cohort Dose/Schedule
6 200 mg 2 weeks on/1 week off
7 200 mg 1 week on/2 weeks off
8 200 mg 2 weeks on/2 weeks off
9 100 mg 2 weeks on/2 weeks off
10 150 mg 1 week on/3 weeks off
11 100 mg QD x 15, then weekly (D1, D8, D15)
12 50 mg QD x 15, then weekly (D1, D8, D15)
= pleural mesothelioma
= non-pleural only mesothelioma
Unaudited data as of 31-July-2023

Preliminary antitumor activity

Case Study 1
- 22 year-old female with advanced dual pleural/peritoneal mesothelioma; no *NF2* mutation
- Prior therapies:
 - cisplatin + pemetrexed + bevacizumab
 - pemetrexed + bevacizumab maintenance
 - ipilimumab + nivolumab
 - PIM kinase inhibitor TP-3654
- Received VT3989 25 mg PO QD, 2 weeks on/2 weeks off; 28-day cycle

Baseline = 16 mm | C14 D1 = 3 mm

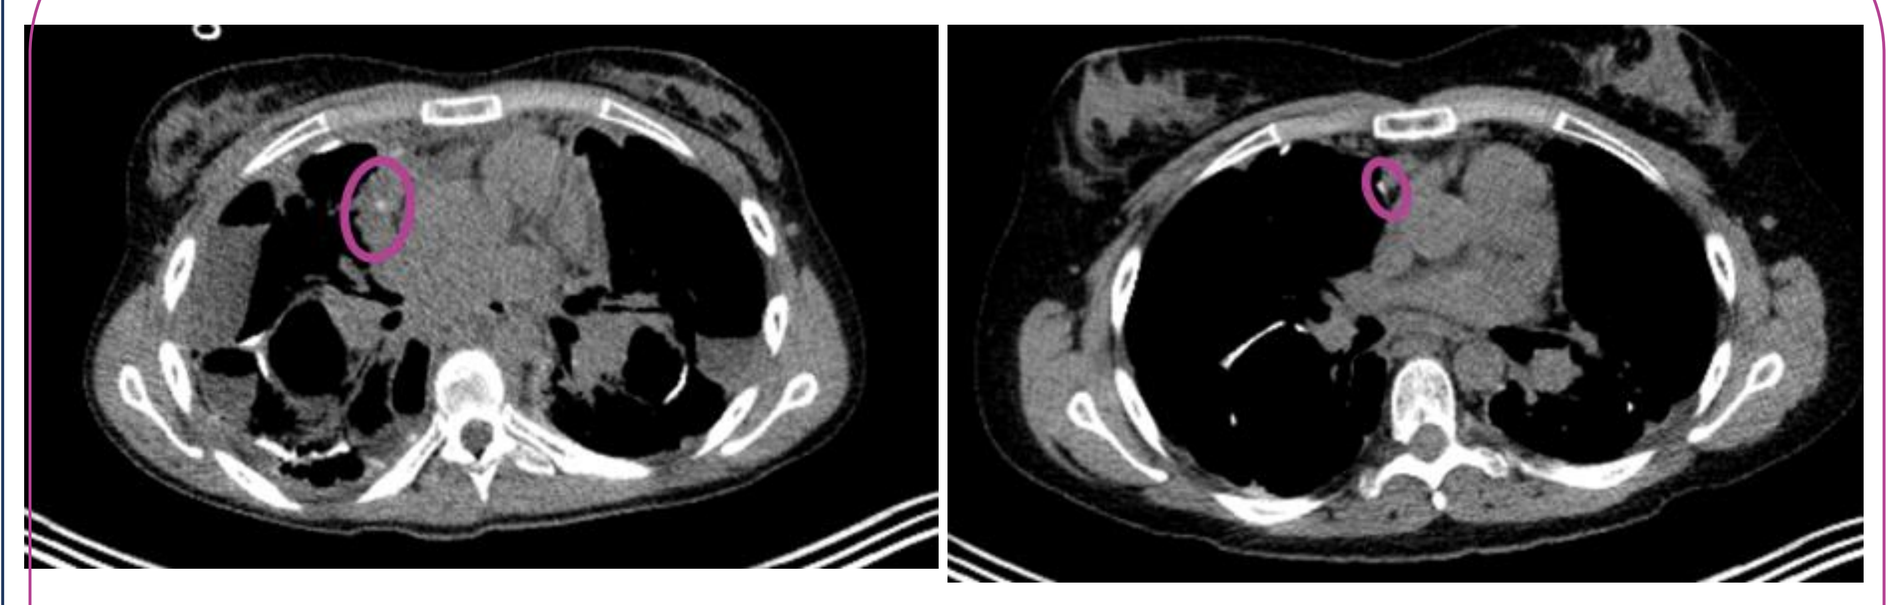

RECIST 1.1 sustained PR (-81.3%)
On study for 17+ months

Case Study 2
- 51 year old male with advanced peritoneal mesothelioma; no *NF2* mutation
- Prior therapies:
 - cisplatin + pemetrexed
 - pembrolizumab
 - carboplatin + pemetrexed
 - pemetrexed maintenance
 - Ipilimumab + Nivolumab
- Received VT3989 50 mg PO QD; 21-day cycle

Baseline = 163 mm | C22 D1 = 100 mm

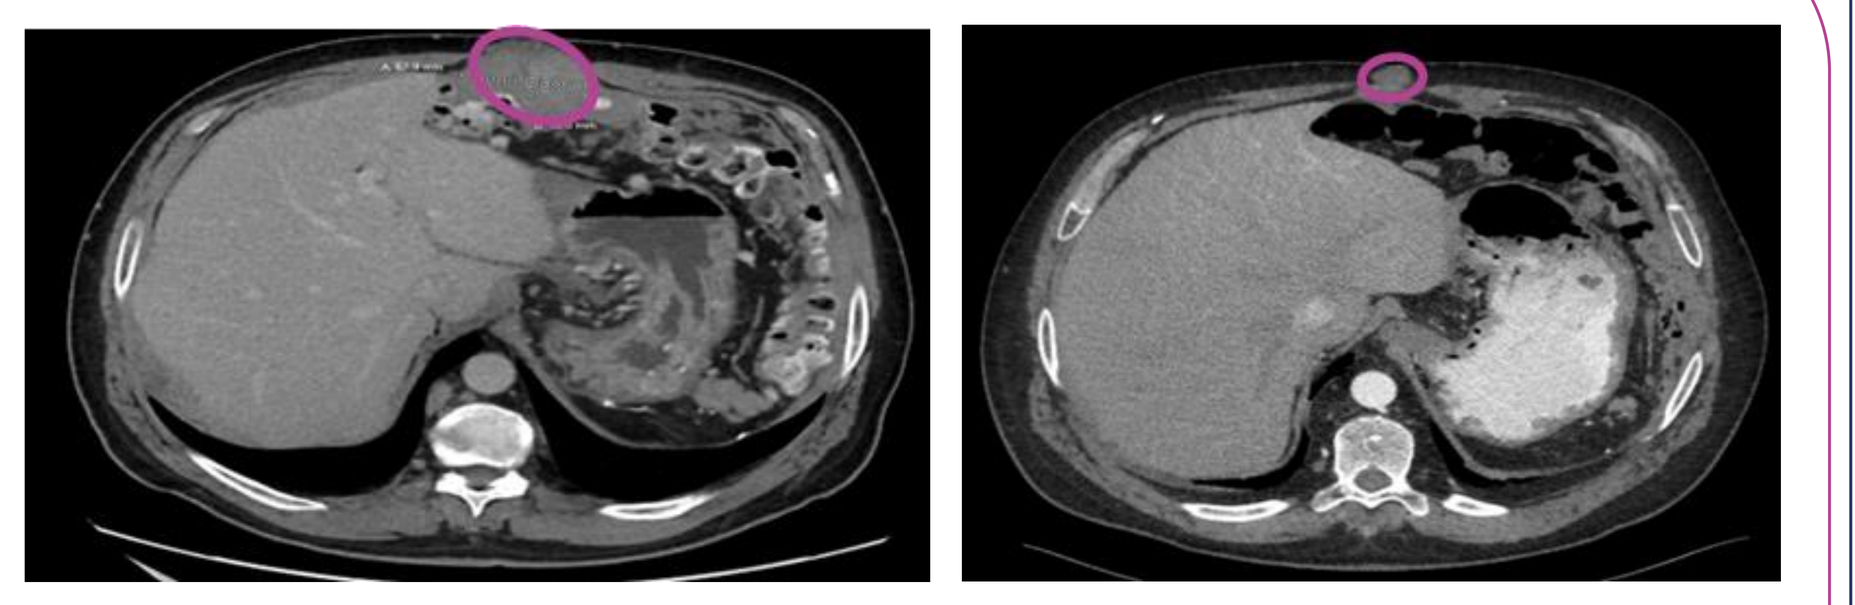

RECIST 1.1 sustained PR (-38.7%)
On study for 25+ months

Presented at: 2023 World Conference on Lung Cancer, September 9-12, Singapore

ClinicalTrials.gov NCT04665206

Contact: F. Andrew Dorr adorr@vivacetherapeutics.com